Cation Exchange Chromatography: An Effective Means of Removing FXIa from a New 10% Intravenous Immunoglobulin Preparation

Alan Huber[a], Gil Bu Kang[b], Min-Jung Kim[b], Seulgi Park[b], Eun Bang[b], Jeungwoon Hong[b], Jihye Lee[b]

[a] GC Biopharma USA, 500 Frank W Burr Blvd, Suite 230, Teaneck, NJ 07666
[b] GCBiopharma, 107 Ihyeon-ro 30-beongil, Giheung-gu, Yongin-si, Gyeonggi-do, Republic of Korea.

INTRODUCTION

- The presence of residual coagulation factors II, VII, IX, X and XIa in intravenous immunoglobulin (IVIG) preparations increases the risk of thrombi formation and thromboembolic events (TEEs) in patients receiving such therapy.[1,2]
- In 2010, a surprising increase in the number of TEEs was observed following use of a 5% IGIV product. Residual FXIa in the product was identified as the root cause.[1]
- IVIG manufacturing via Cohn-Oncley cold ethanol fractionation has been demonstrated to successfully remove FII, FVII, FIX, and FX.[3,4]
- Ethanol precipitation does not effectively separate FXIa from IVIG due to the high isoelectric point of FXIa (8.9 - 9.1).[5]
- FXIa can remain in intermediary products, risking the presence of procoagulant activity in the final IVIG preparation.
- The development of processes to remove residual FXIa from IVIG has been a longstanding goal with the aim of improving product safety.

STUDY OBJECTIVES

- To develop and evaluate a process to remove FXIa from the GC Biopharma 10% IVIG (GC5107) preparation.
- To assess the robustness of the process via a spiking study.

GC5107 MANUFACTURING PROCESS

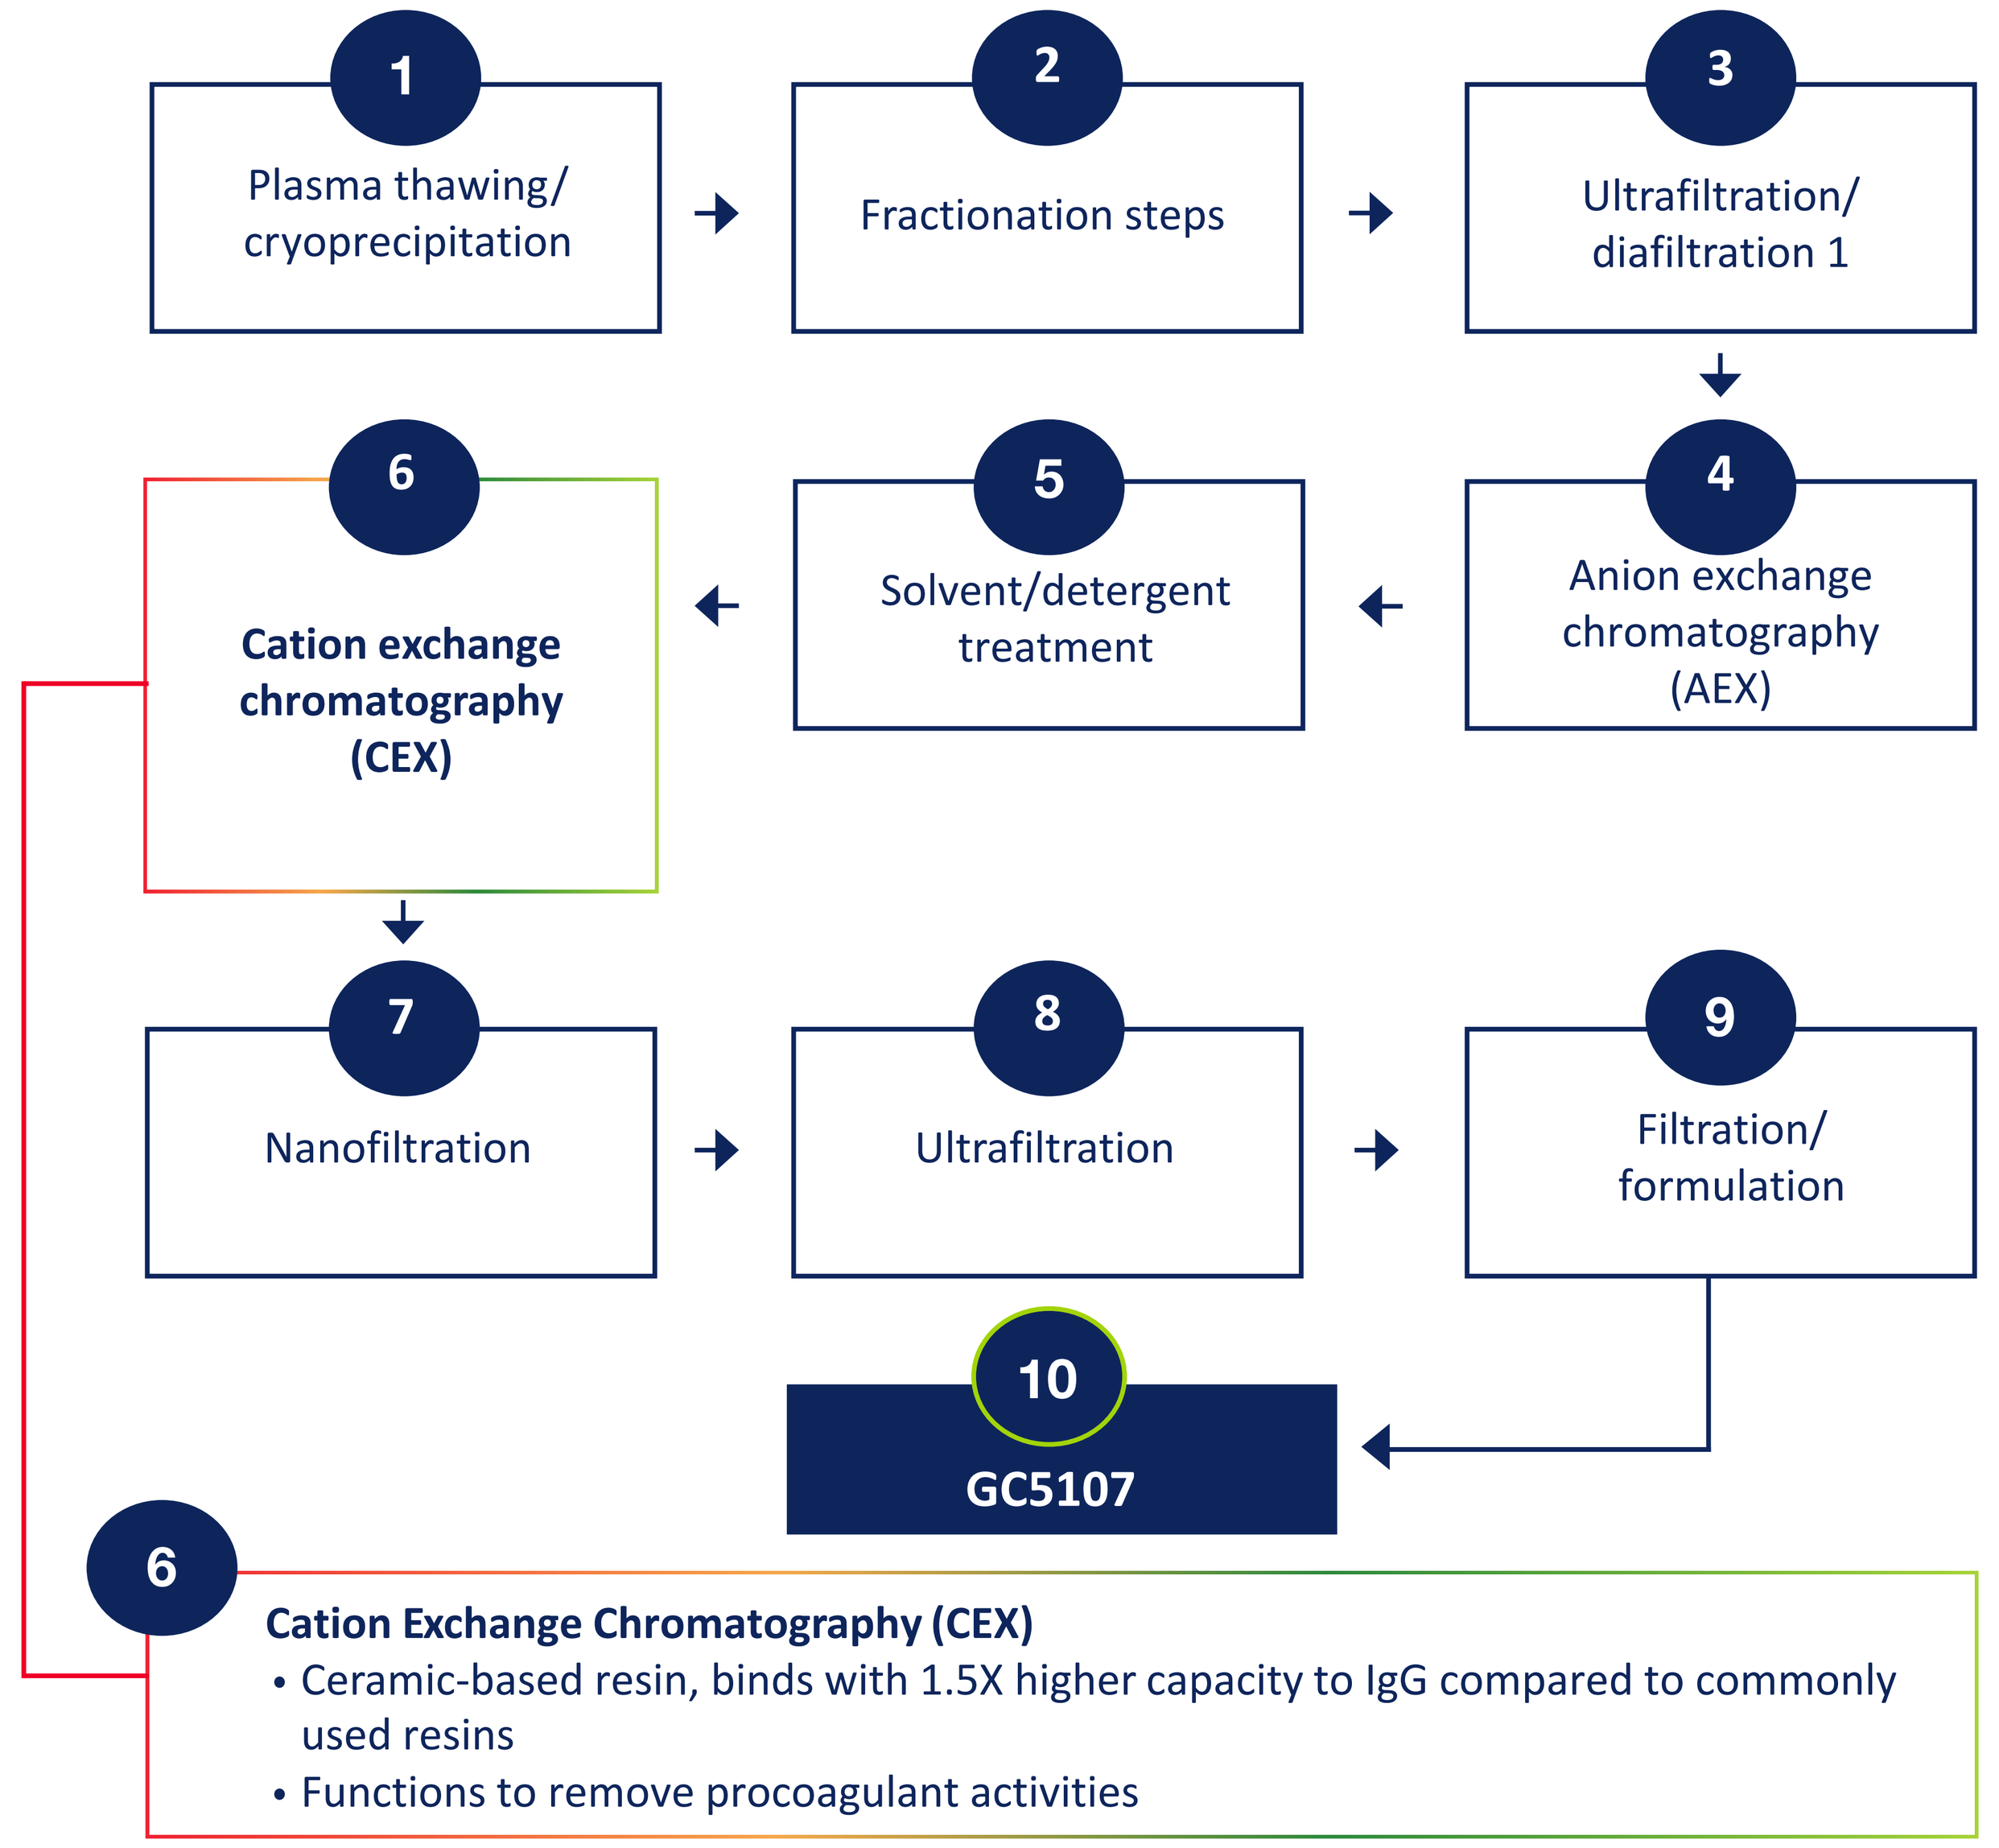
1 Plasma thawing/ cryoprecipitation
2 Fractionation steps
3 Ultrafiltration/ diafiltration 1
4 Anion exchange chromatography (AEX)
5 Solvent/detergent treatment
6 Cation exchange chromatography (CEX)
7 Nanofiltration
8 Ultrafiltration
9 Filtration/ formulation
10 GC5107
6 Cation Exchange Chromatography (CEX)
• Ceramic-based resin, binds with 1.5X higher capacity to IgG compared to commonly used resins
• Functions to remove procoagulant activities

METHODS

Procoagulant activity in intermediate and final purification products of the GC5107 manufacturing process was assessed using:

- Thrombin generation assay (TGA)
- Chromogenic FXIa assay
- Non-activated partial thromboplastin time (NaPTT)
- FXI/FXIa ELISA
- Western blot analysis

RESULTS

INCORPORATING CEX RESULTS IN LOW LEVELS OF PROCOAGULANT ACTIVITY IN THE FINAL PRODUCT

Batch no.	TGA (mIU/mL)	Chromogenic FXIa (mIU/mL)	NaPTT (s)			
			1:5	Ratio	1:10	Ratio
1	<1.56	<0.16	267.0	1.0	266.2	1.0
2	<1.56	<0.16	282.6	1.1	268.8	1.1
3	<1.56	<0.16	268.1	1.0	275.6	1.1
4	<1.56	<0.16	299.4	1.0	291.9	1.0
5	<1.56	<0.16	302.4	1.0	304.8	1.0
6	<1.56	<0.16	300.8	1.0	296.6	1.0
7	<1.56	<0.16	265.9	1.1	265.9	1.1
8	<1.56	<0.16	256.8	1.1	259.6	1.0
9	<1.56	<0.16	271.3	1.1	266.7	1.1
Mean ± SD	<1.56	<0.16	279.4 ± 17.5	1.0 ± 0.1	277.3 ± 16.2	1.0 ± 0.1

Table 1. Procoagulant activity in 9 lots of GC5107.

INCORPORATING CEX REDUCES FXIa TO BELOW DETECTION LIMITS

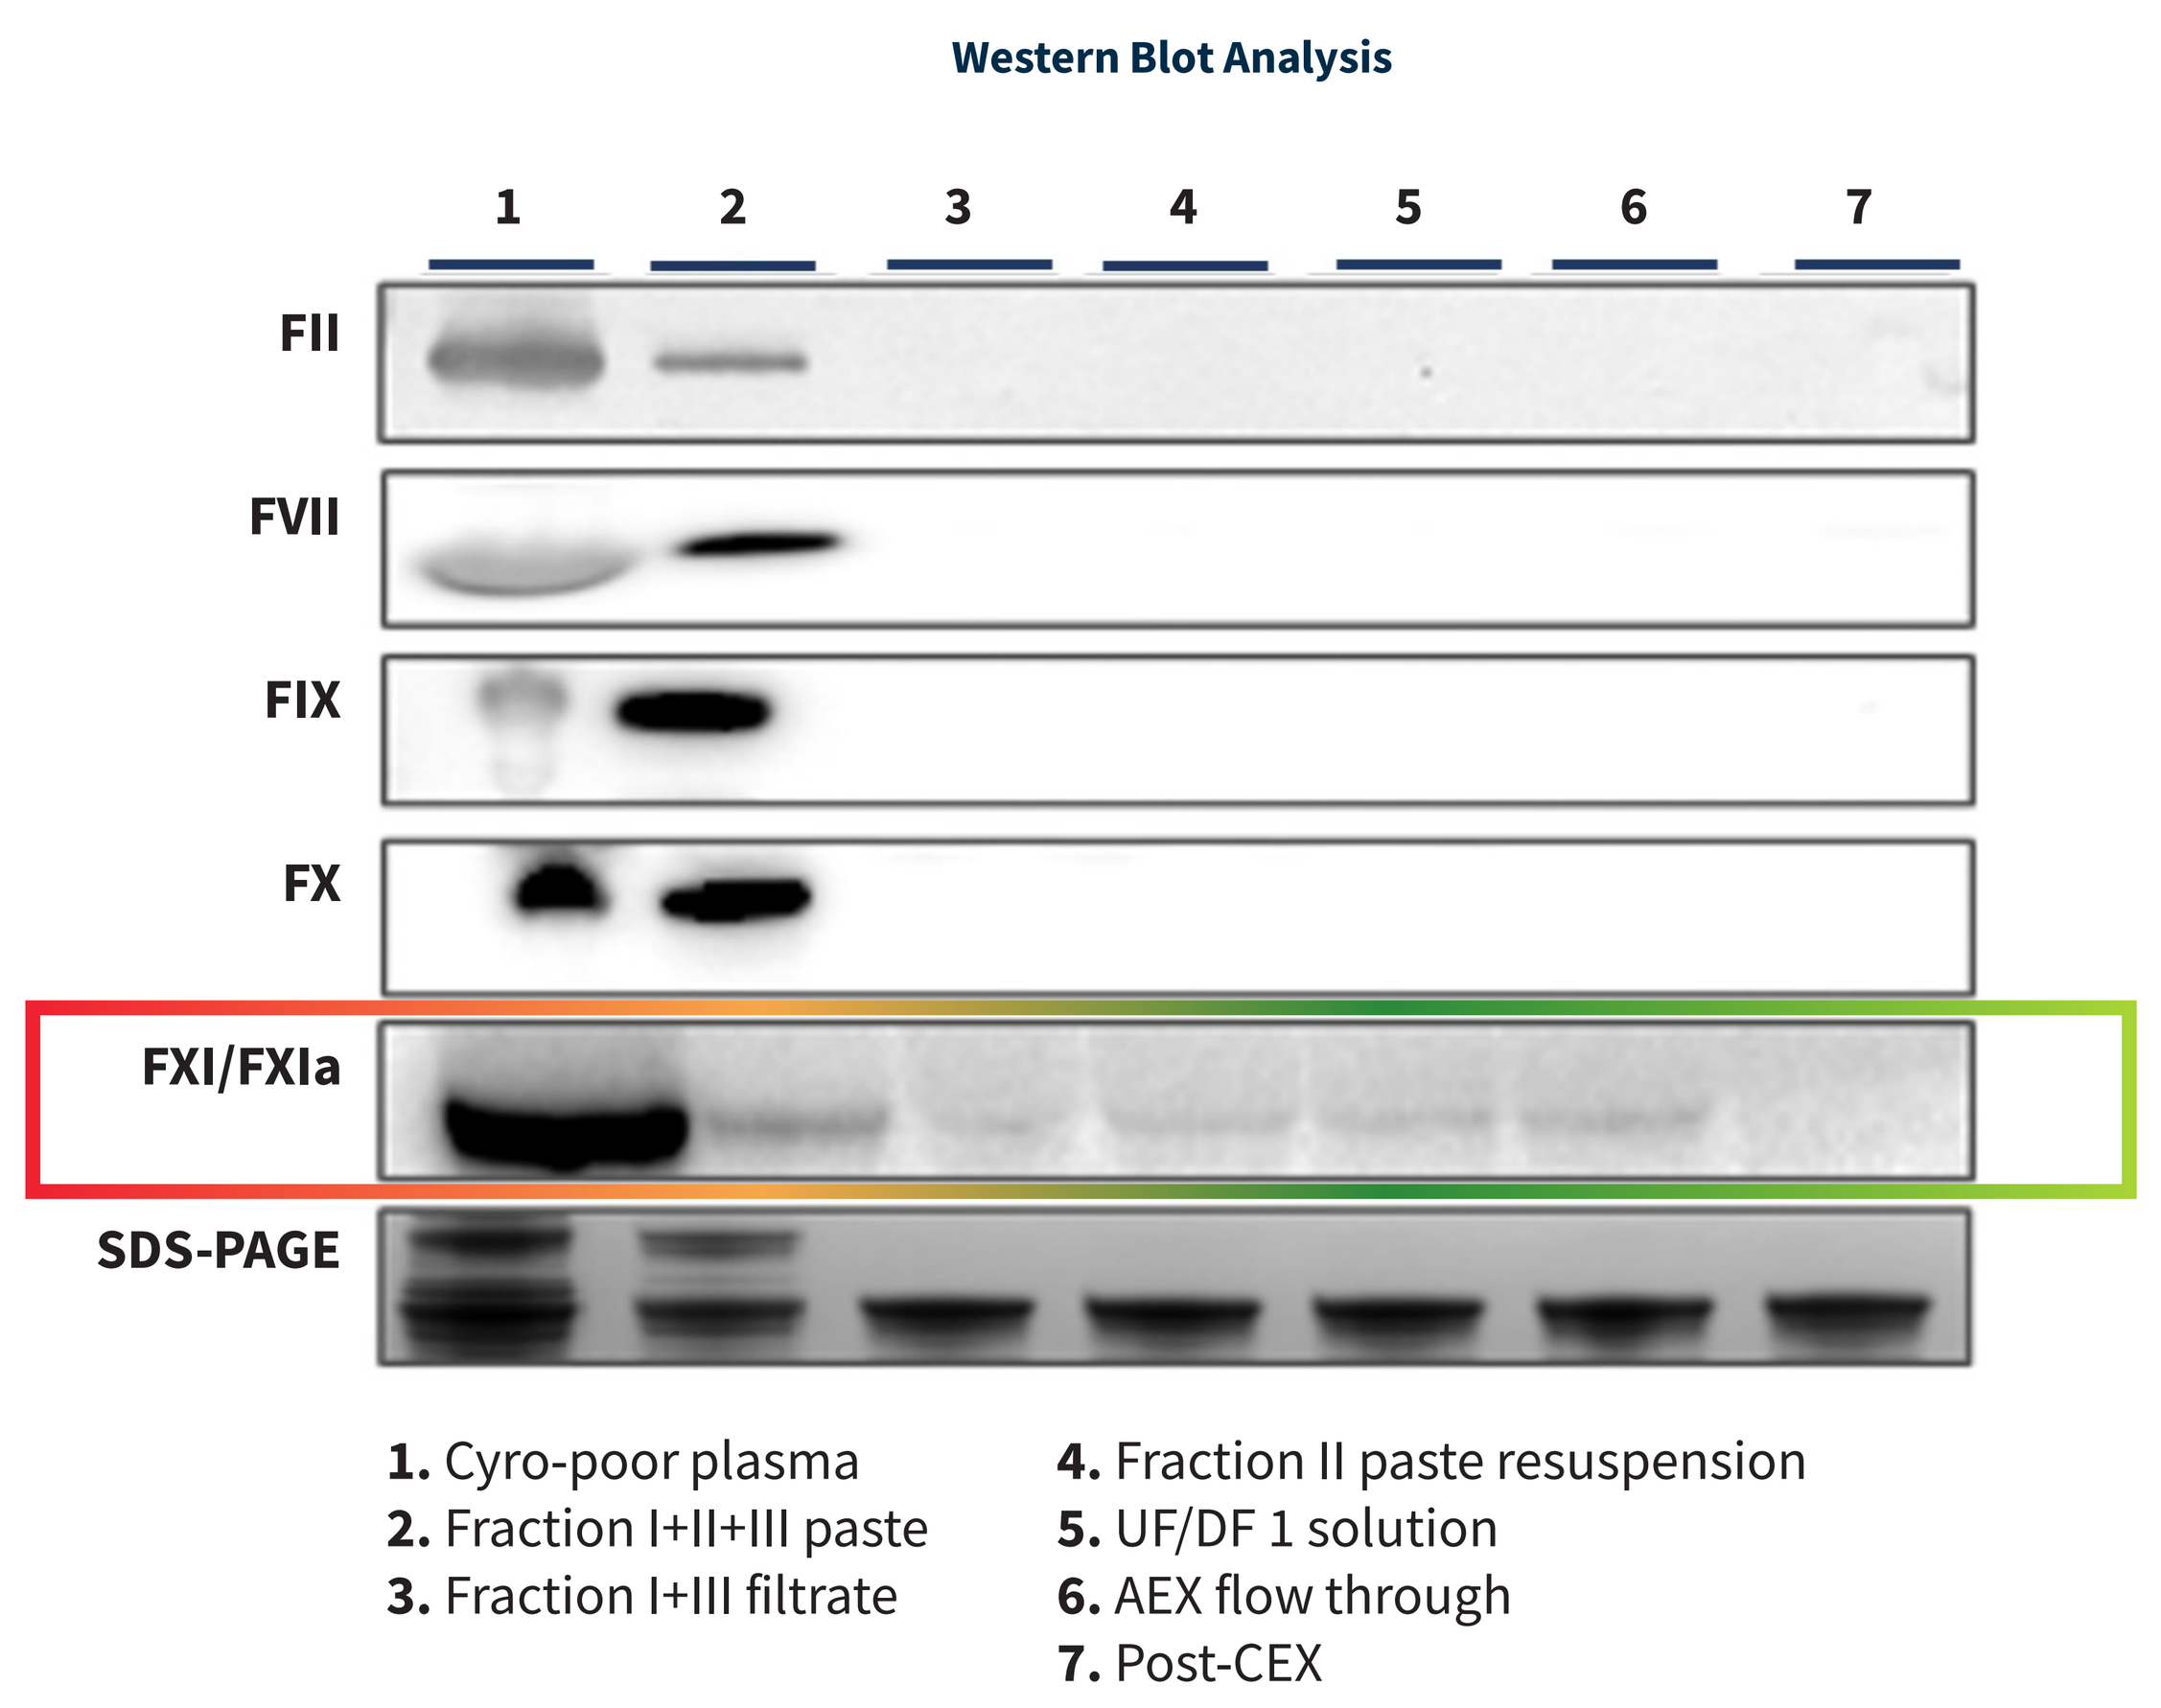
Western Blot Analysis
1 2 3 4 5 6 7
FII
FVII
FIX
FX
FXI/FXIa
SDS-PAGE
1. Cyro-poor plasma
2. Fraction I+II+III paste
3. Fraction I+III filtrate
4. Fraction II paste resuspension
5. UF/DF 1 solution
6. AEX flow through
7. Post-CEX

Figure 1. Western Blot analysis of coagulation factors (with SDS-PAGE loading control) show removal of FII, FVII, FIX and FX after cold ethanol fractionation. FXIa is removed only following CEX chromatography.

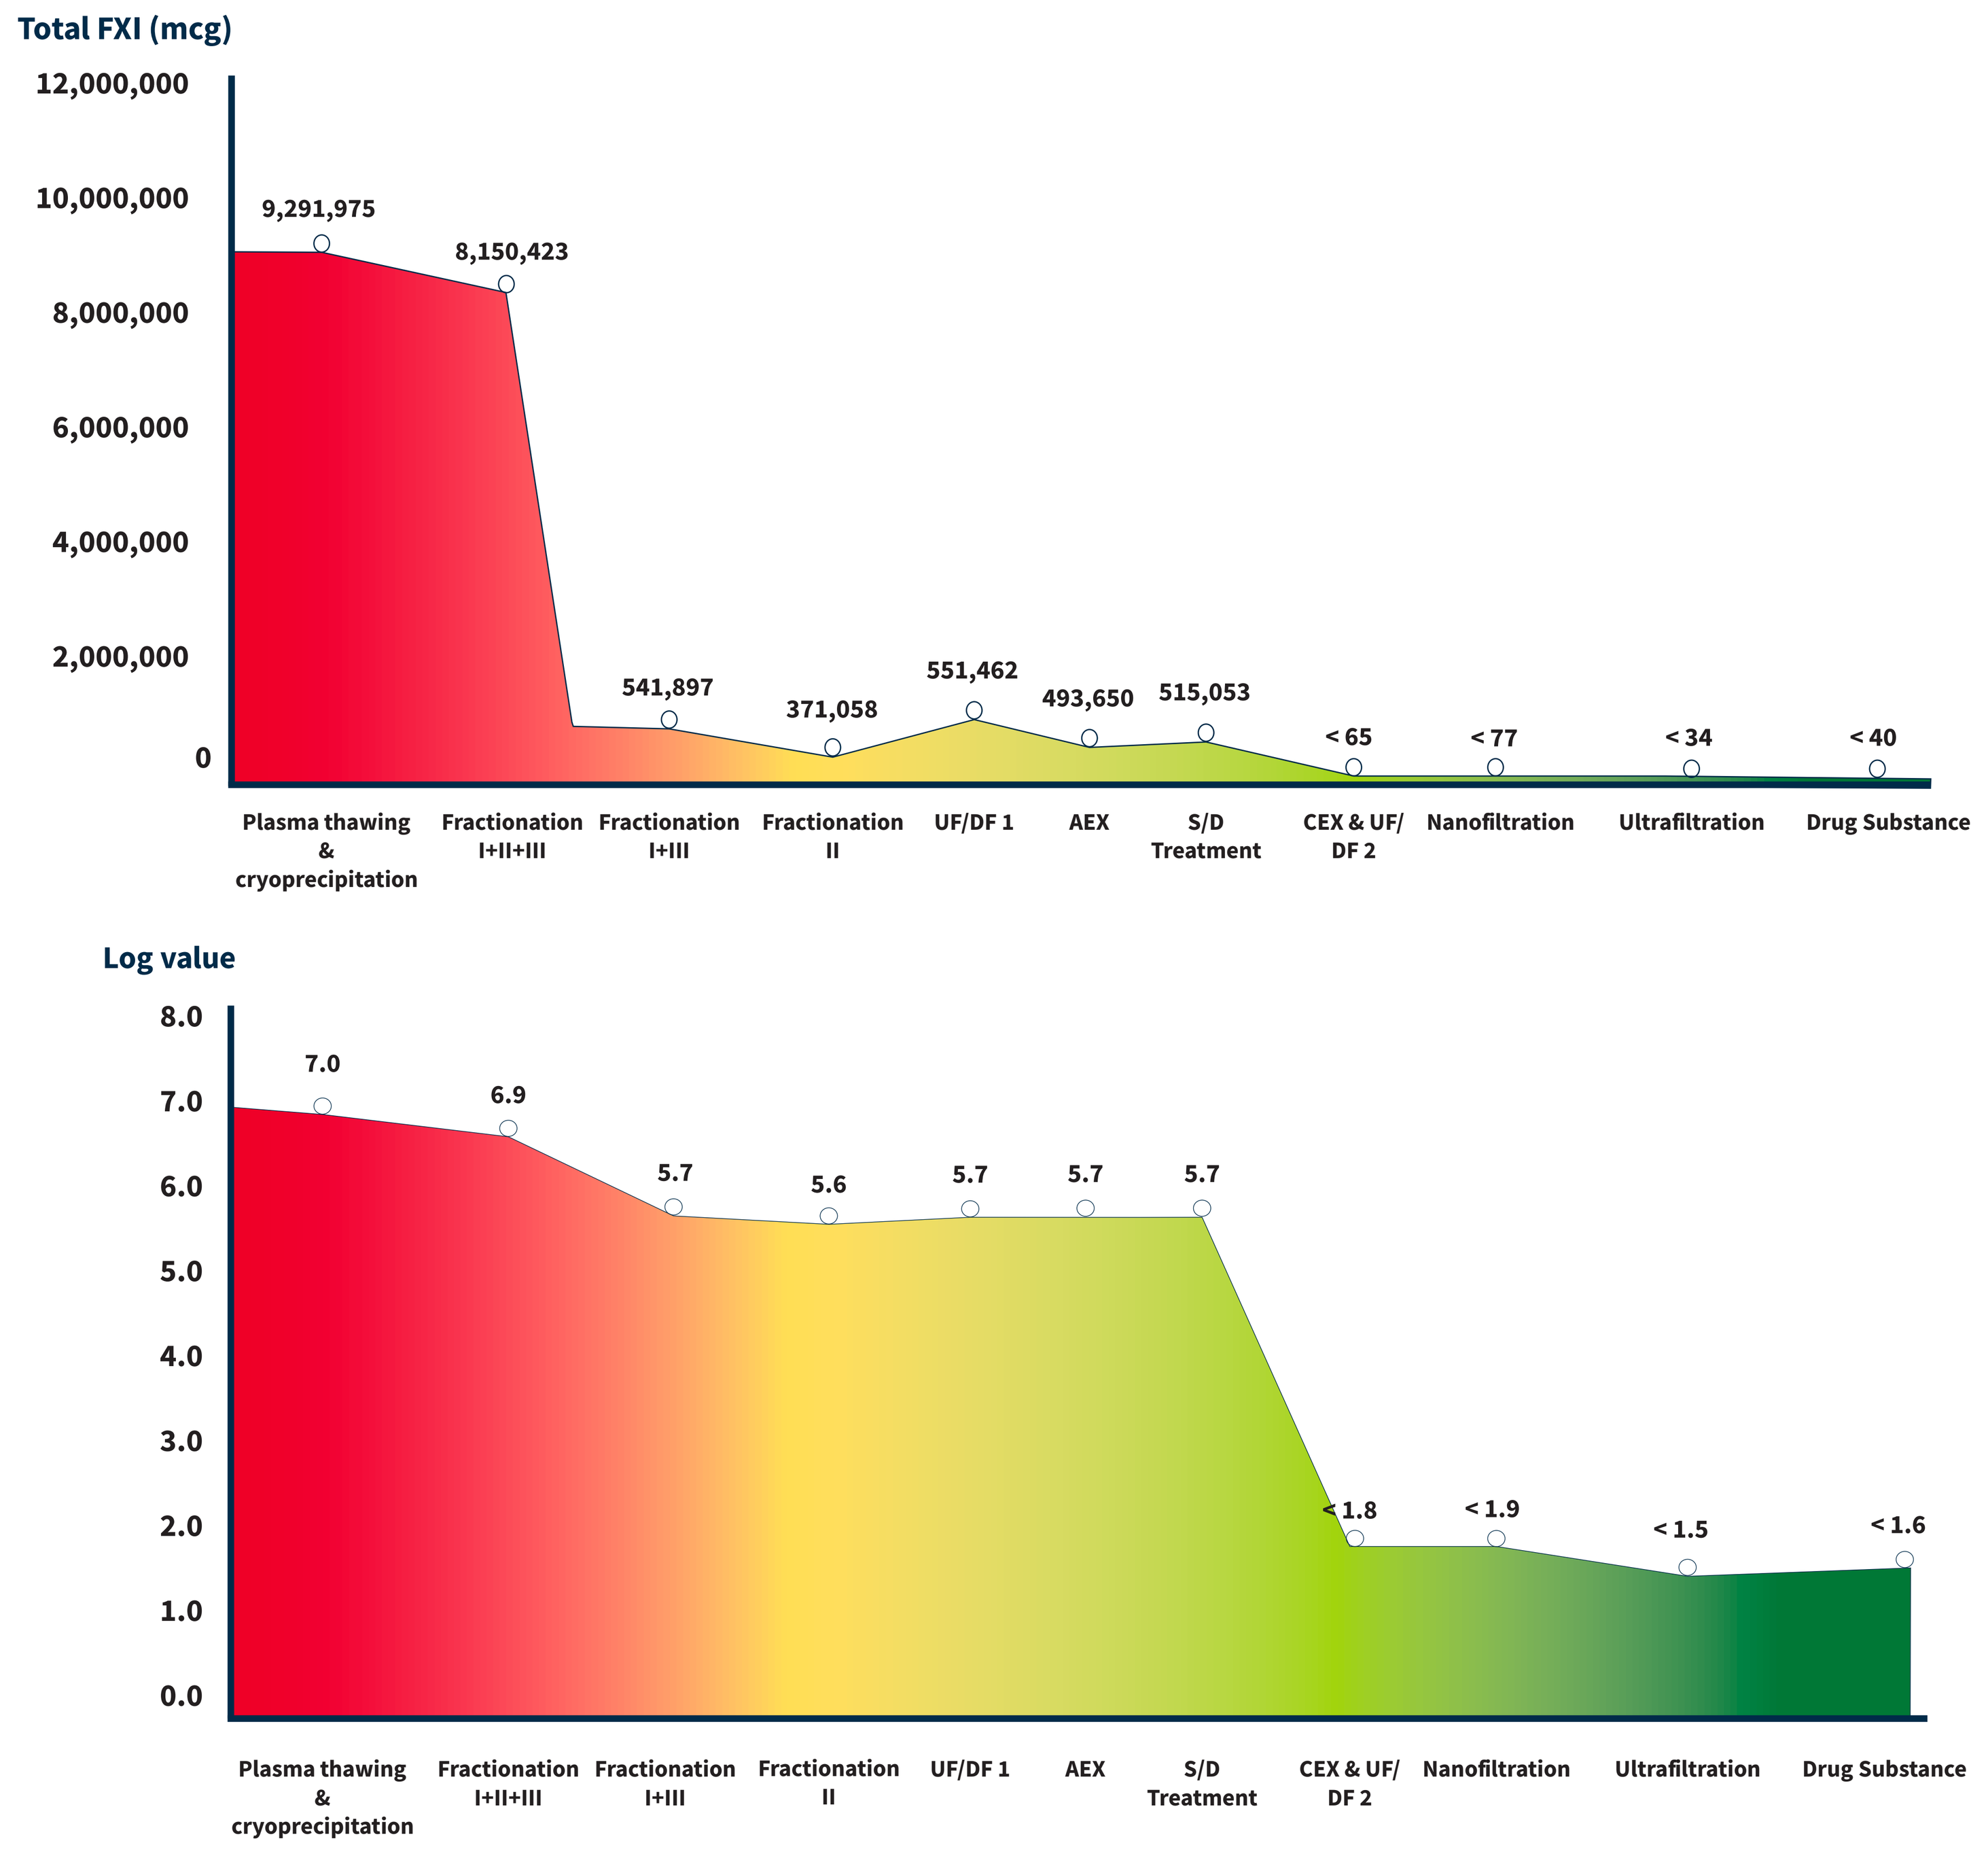
Total FXI (mcg)
12,000,000
10,000,000
8,000,000
6,000,000
4,000,000
2,000,000
0
9,291,975
8,150,423
541,897
371,058
551,462
493,650
515,053
< 65
< 77
< 34
< 40
Plasma thawing & cryoprecipitation
Fractionation I+II+III
Fractionation I+III
Fractionation II
UF/DF 1
AEX
S/D Treatment
CEX & UF/ DF 2
Nanofiltration
Ultrafiltration
Drug Substance
Log value
8.0
7.0
6.0
5.0
4.0
3.0
2.0
1.0
0.0
7.0
6.9
5.7
5.6
5.7
5.7
5.7
< 1.8
< 1.9
< 1.5
< 1.6
Plasma thawing & cryoprecipitation
Fractionation I+II+III
Fractionation I+III
Fractionation II
UF/DF 1
AEX
S/D Treatment
CEX & UF/ DF 2
Nanofiltration
Ultrafiltration
Drug Substance

Figure 2. Analysis of total FXI levels with the CEX chromatography step highlighted.
CEX chromatography reduced total FXI from ~160,000mg to < 68mg.

SPIKING STUDY DEMONSTRATES ROBUST ABILITY OF CEX TO REMOVE FXIa

FXIa content	No spike				FXIa spike (4.0 µg/mL)				FXIa spike (21.0 µg/mL)			
	Pre-CEX	Post-CEX	IgG recovery (%)	Residual	Pre-CEX	Post-CEX	IgG recovery (%)	Residual	Pre-CEX	Post-CEX	IgG recovery (%)	Residual
Protein [ELISA] (ng/mL)	**83.79**	**< 0.31**	103.3	NC*	**2679.48**	**0.96**	100.8	0.01	**13264.79**	**5.56**	102.4	0.01
Activity [TGA] (mU/mL)	**22.26**	**< 1.56**		NC	**73159.54**	**< 1.56**		NC	**367549.59**	**< 1.56**		NC

*NC: Not calculated

Table 2. Results of spiking study in which starting samples were spiked with FXIa at a concentration of 4.0 mg/mL and 21.0 mg/mL (representing concentrations 32.5X and 169.9X higher than that found in a normal specimen.[6])

DISCUSSION

- The GC5107 manufacturing process including CEX chromatography reduced procoagulant activity in the final product to below detection limits (Table 1) as measured by[6]:
 - TGA assay (< 1.56 mIU/mL)
 - Chromogenic FXIa assay (< 0.16 mIU/mL)
- The NaPTT of >250 sec in all 9 batches of GC5107 indicates very low levels of procoagulant activity (Table 1).
- Western blot analysis of intermediate samples indicated that FII, FVII, FIX, and FX were removed through cold ethanol fractionation, while FXIa was successfully reduced to undetectable levels only after CEX chromatography (Figures 1 and 2).
- In starting samples spiked with FXIa at 32.5X and 169.9X levels found in normal samples, the GC5107 manuzfacturing process reduced FXIa to very low levels (by ELISA) and reduced procoagulant activity to below the detection limit (by TGA) (Table 2).

CONCLUSION

- These results demonstrate the ability of CEX chromatography to effectively remove FXIa (by Western blot), and reduce procoagulant activity to below detection limits (by TGA and chromogenic FXIa assay).
- The results of the spiking study demonstrate the robustness of the process — even for starting material containing 32X and 169.9X the FXIa content of a normal sample.
- The new manufacturing process including CEX chromatography may serve to reduce the risk of TEEs in patients receiving this therapy.

ABBREVIATIONS

CEX: Cation exchange chromatography
ELISA: Enzyme-linked immunosorbent assay IVIG: Intravenous immunoglobulin
NaPTT: Non-activated partial thromboplastin time
NC: Not calculated
TEEs: Thromboembolic events
TGA: Thrombin generation assay
SD: Standard deviation

REFERENCES

1. Roemisch JR, Kaar W, Zoechling A, et al. Identification of activated FXI as the major biochemical root cause in IVIG batches associated with thromboembolic events. Analytical and experimental approaches resulting in corrective and preventive measures implemented into the Octagam® Manufacturing process. WebmedCentral 2011;2. WMC002002. 2. Wolberg AS, Kon RH, Monroe DM, Hoffman M. Coagulation factor FXI is a contaminant in intravenous immunoglobulin preparations. *Am J Hematol.* 2000;65:30-4. 3. Cohn EJ, Strong LE, Hughes Jr WL, et al. Preparation and properties of serum and plasma proteins III. A system for the separation into fractions of the protein and lipoprotein components of biological tissues and fluids. *J Am Chem Soc.* 1946;68:459-75. 4. Oncley JL, Melin M, Richert DA, Cameron JW, Gross Jr PM. The separation of the antibodies, isoagglutinins, prothrombin, plasminogen and beta1-lipoprotein into subfractions of human plasma. J *Am Chem Soc.* 1949;71: 541-50. 5. Burnouf-Radosevich M, Burnouf T. A therapeutic, highly purified factor XI concentrate from human plasma. *Transfusion*. 1992;32:861-7. 6. Park DH, Kang GB, Kang DE, et al. A new manufacturing process to remove thrombogenic factors (II, VII, IX, X, and XI) from intravenous immunoglobulin preparations. *Biologicals*. 2017;45:1-8.

GC Biopharma USA

Previously published in Frontiers of Cardiovascular Medicine, November 22, 2023. Kang GB, Huber A, Lee J, Kim M-J, Bang E, Hong J and Park S (2023) Cation exchange chromatography removes FXIa from a 10% intravenous immunoglobulin preparation. Front. Cardiovasc. Med. 10:1253177. doi: 10.3389/fcvm.2023.1253177. Encore presentation at National Association of Specialty Pharmacy (NASP) Annual Meeting, October 6-9, 2024, Nashville, TN.