Comparative Effectiveness of Denosumab versus Zoledronic Acid among Postmenopausal Women with Osteoporosis in the U.S. Medicare Program

THE UNIVERSITY OF ALABAMA AT BIRMINGHAM

Jeffrey R. Curtis[1,2], Tarun Arora[2], Ye Liu[1], Tzu-Chieh Lin[3], Leslie Spangler[3], Vanessa C. Brunetti[3], Robert K. Stad[3], Michele McDermott[3], Brian D. Bradbury[3], Min Kim[3]

[1] University of Alabama at Birmingham, Birmingham, AL, USA; [2] Foundation for Advancing Science, Technology, Education and Research, Birmingham, AL, USA; [3] Amgen Inc., Thousand Oaks, CA, USA.

faster

BACKGROUND

Although clinical trials have shown that denosumab (Dmab) significantly increases bone mineral density at key skeletal sites more than zoledronic acid (ZA), evidence from randomized trials evaluating fracture outcomes is lacking.

OBJECTIVES

This retrospective cohort study evaluated the comparative effectiveness of Dmab versus ZA in reducing fracture risk among women with postmenopausal osteoporosis (PMO) in the U.S.

METHODS

Female Medicare fee-for-service beneficiaries ≥ 66 years of age who newly initiated Dmab (n=89,990) or ZA (n=36,861) between Jan 1, 2012 to Dec 31, 2018 with no prior history of osteoporosis treatment were followed from treatment initiation (index date) until the first instance of a given fracture outcome, treatment discontinuation (defined as the end of exposure according to usual dosing intervals + 60-day gap) or switch, Medicare disenrollment, death, end of available data (Dec 31, 2019), or 5 years post-index date. A doubly robust inverse-probability of treatment (weights estimated from multivariate logistic regression models) and censoring (weights estimated from multivariate Cox Proportional Hazards regression models) weighted function was used to estimate the relative risk (RR) associated with the use of Dmab compared with ZA for major osteoporotic (MOP; nonvertebral and hospitalized vertebral), hip, nonvertebral (NV; includes hip, humerus, pelvis, radius/ulna, other femur), non-hip, nonvertebral (NHNV), and hospitalized vertebral (HV) fractures for the overall study period and by year of follow-up.

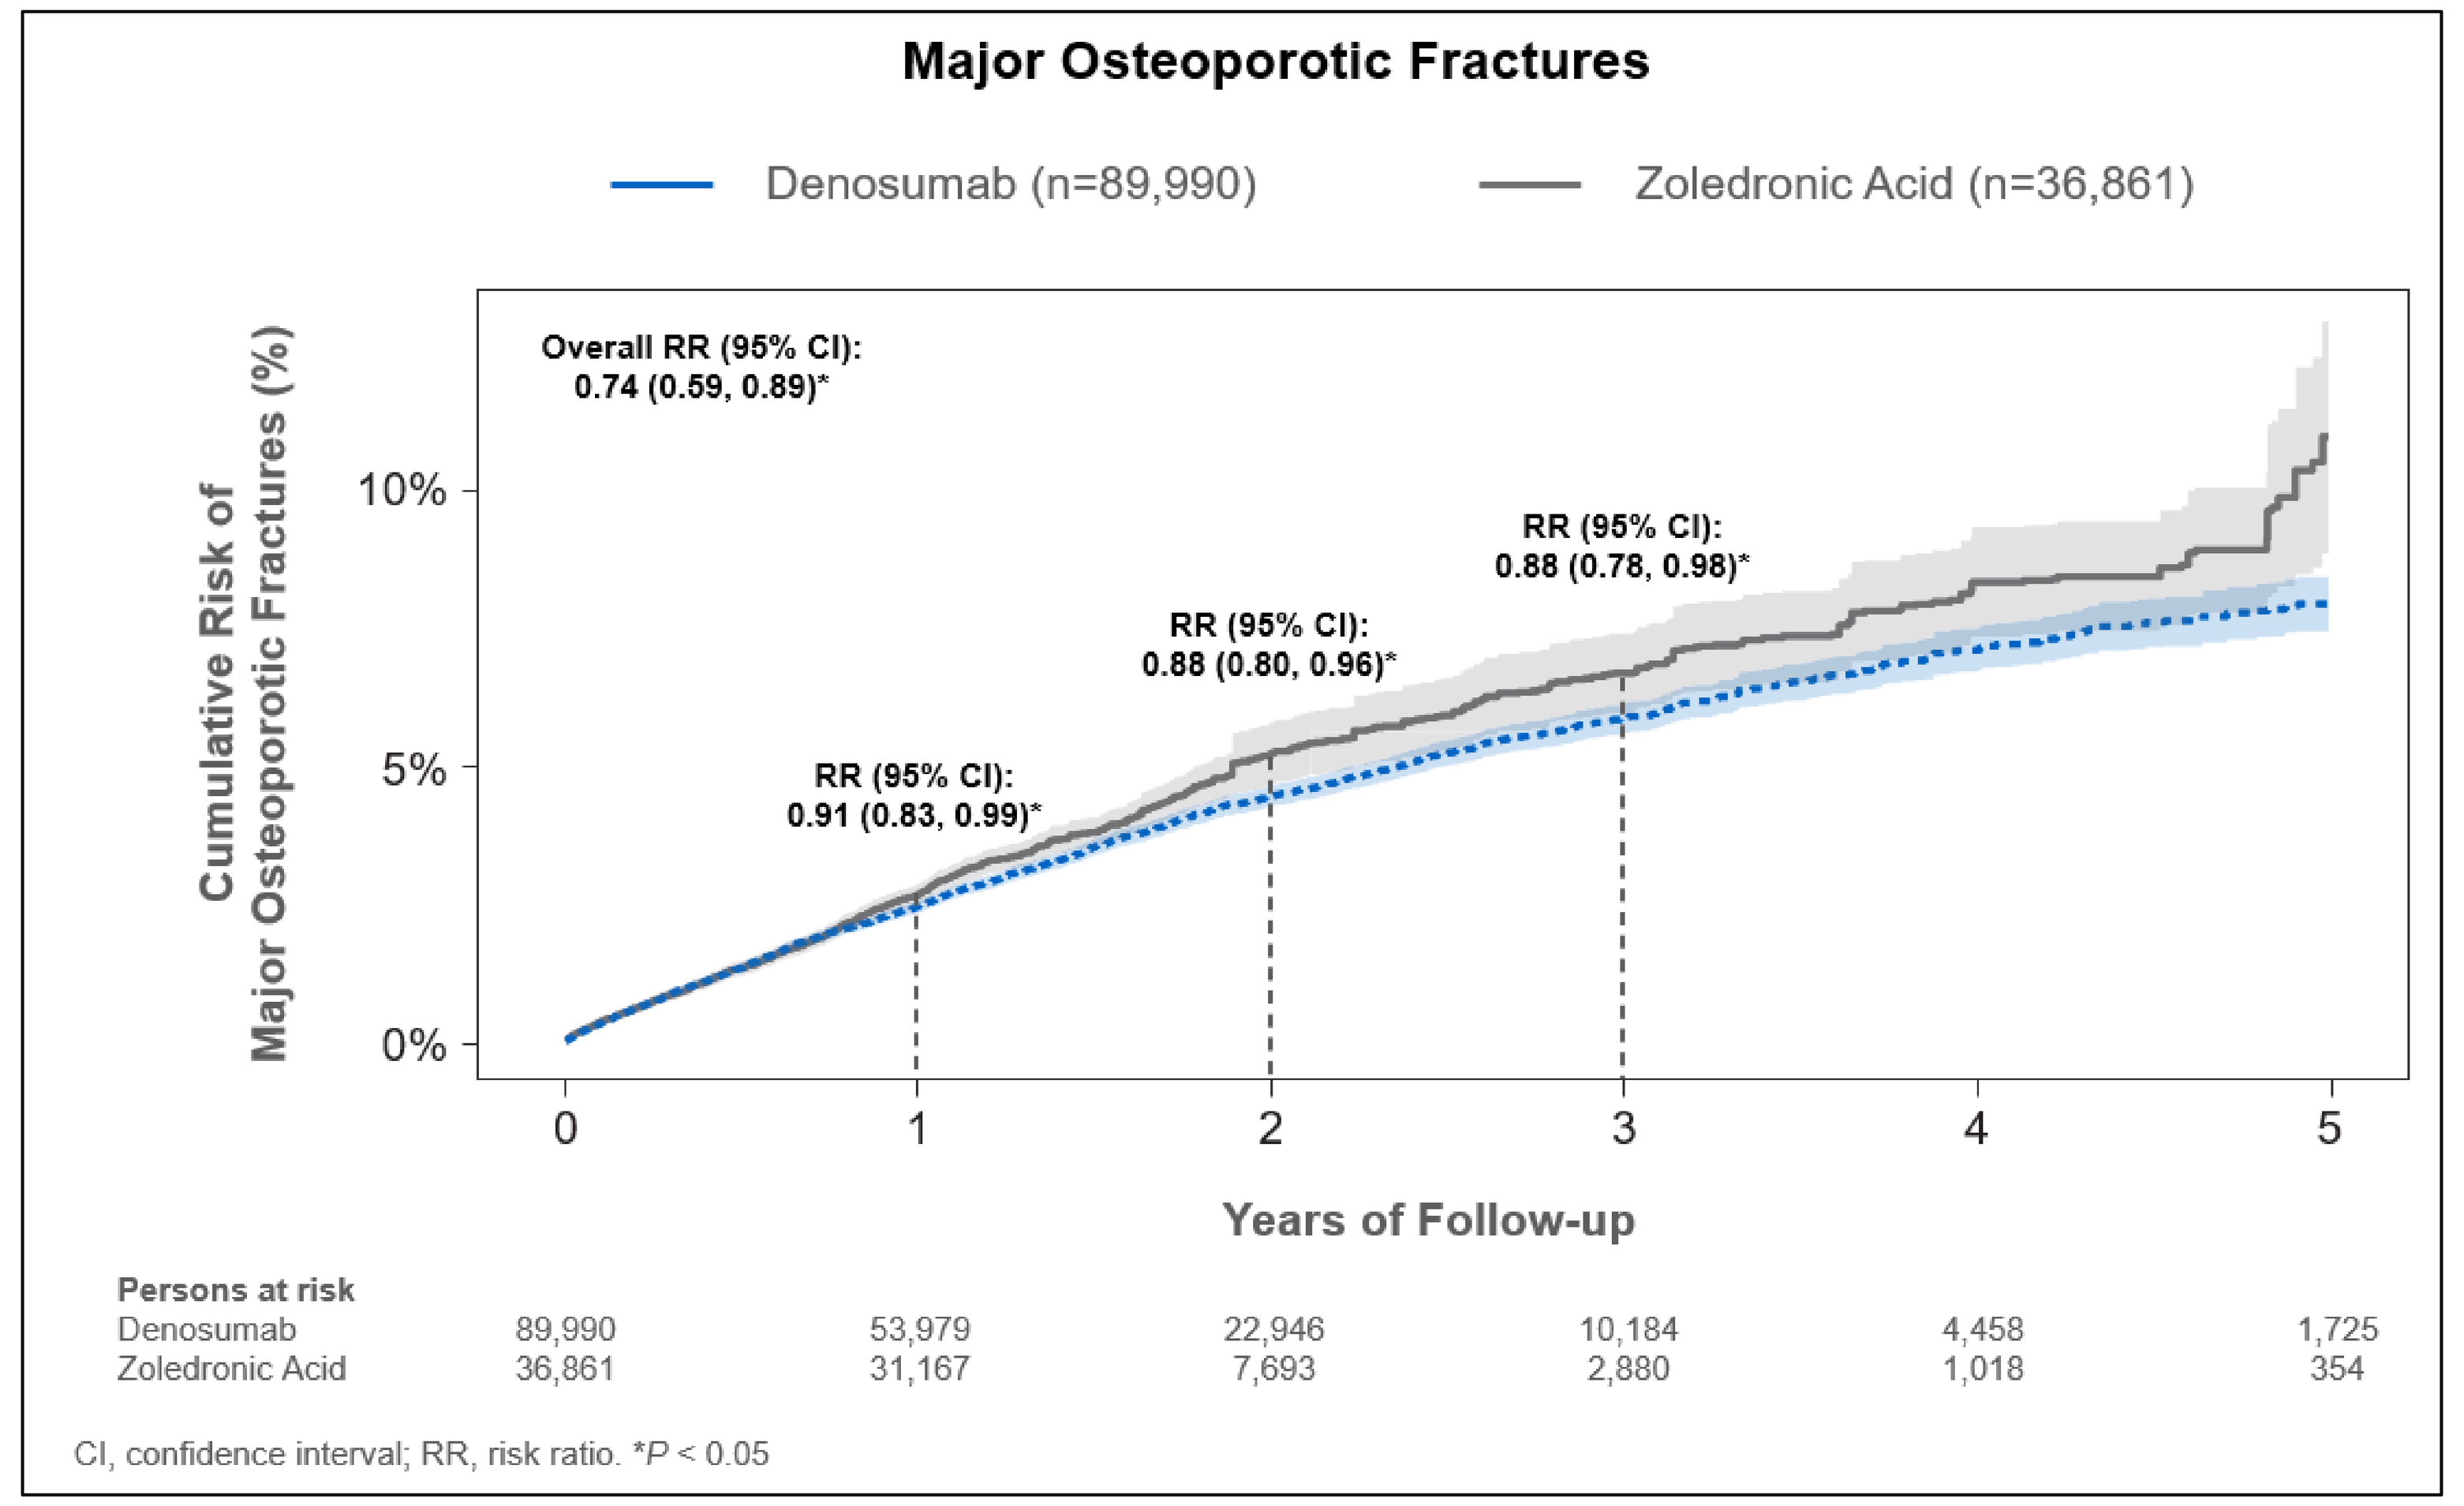
Major Osteoporotic Fractures
Denosumab (n=89,990)
Zoledronic Acid (n=36,861)
Cumulative Risk of Major Osteoporotic Fractures (%)
Overall RR (95% CI): 0.74 (0.59, 0.89)*
RR (95% CI): 0.91 (0.83, 0.99)*
RR (95% CI): 0.88 (0.80, 0.96)*
RR (95% CI): 0.88 (0.78, 0.98)*
Years of Follow-up
Persons at risk
Denosumab 89,990 53,979 22,946 10,184 4,458 1,725
Zoledronic Acid 36,861 31,167 7,693 2,880 1,018 354
CI, confidence interval; RR, risk ratio. *P < 0.05

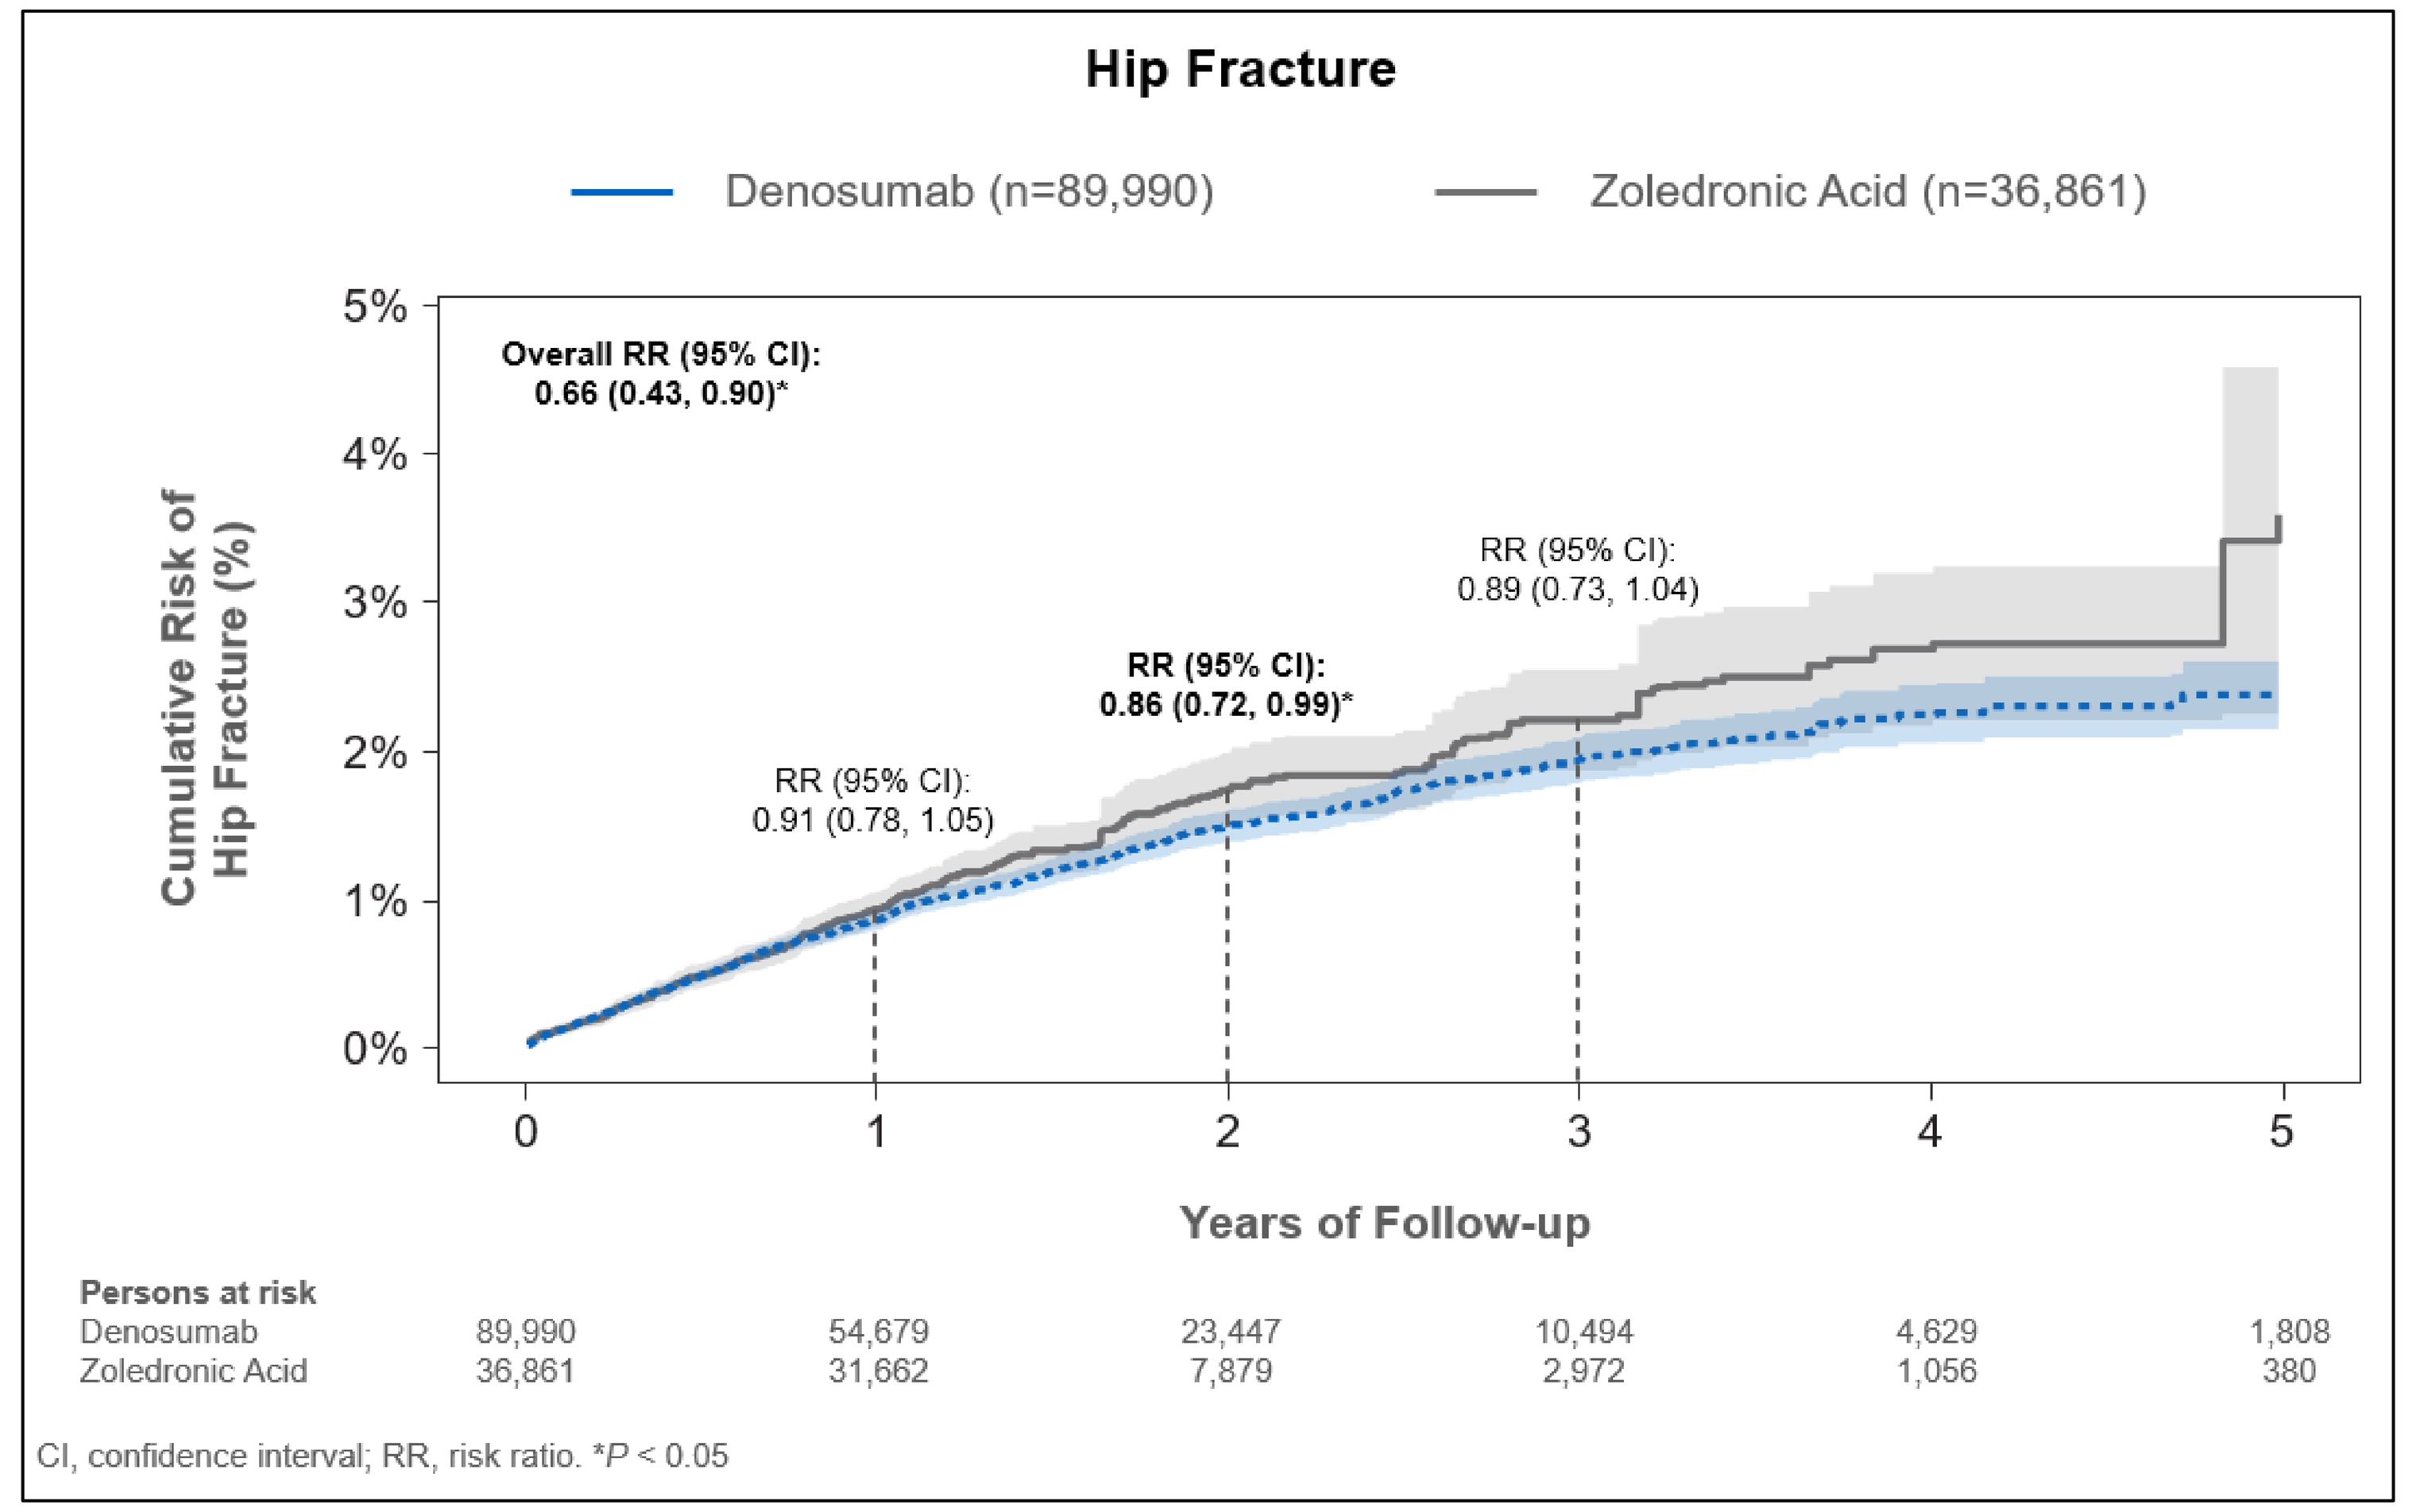
Hip Fracture
Denosumab (n=89,990)
Zoledronic Acid (n=36,861)
Cumulative Risk of Hip Fracture (%)
Overall RR (95% CI): 0.66 (0.43, 0.90)*
RR (95% CI): 0.91 (0.78, 1.05)
RR (95% CI): 0.86 (0.72, 0.99)*
RR (95% CI): 0.89 (0.73, 1.04)
Years of Follow-up
Persons at risk
Denosumab 89,990 54,679 23,447 10,494 4,629 1,808
Zoledronic Acid 36,861 31,662 7,879 2,972 1,056 380
CI, confidence interval; RR, risk ratio. *P < 0.05

RESULTS

Over a maximum of 5 years of follow-up, Dmab reduced the risk of MOP by 26% (RR=0.74; 95% CI: 0.59-0.89), hip by 34% (0.66; 0.43-0.90), NV by 33% (0.67; 0.52-0.82), and NHNV by 31% (0.69; 0.50-0.88), and HV fractures by 10% (0.90; 0.56-1.24) compared with ZA. Over time, Dmab reduced the risk of MOP fractures by 9% (0.91; 0.83-0.99) at year 1, 12% (0.88; 0.80-0.96) at year 2, and 12% (0.88; 0.78-0.98) at year 3. An increase in the magnitude of fracture risk reduction with increasing duration of exposure was also observed for other NV outcomes.

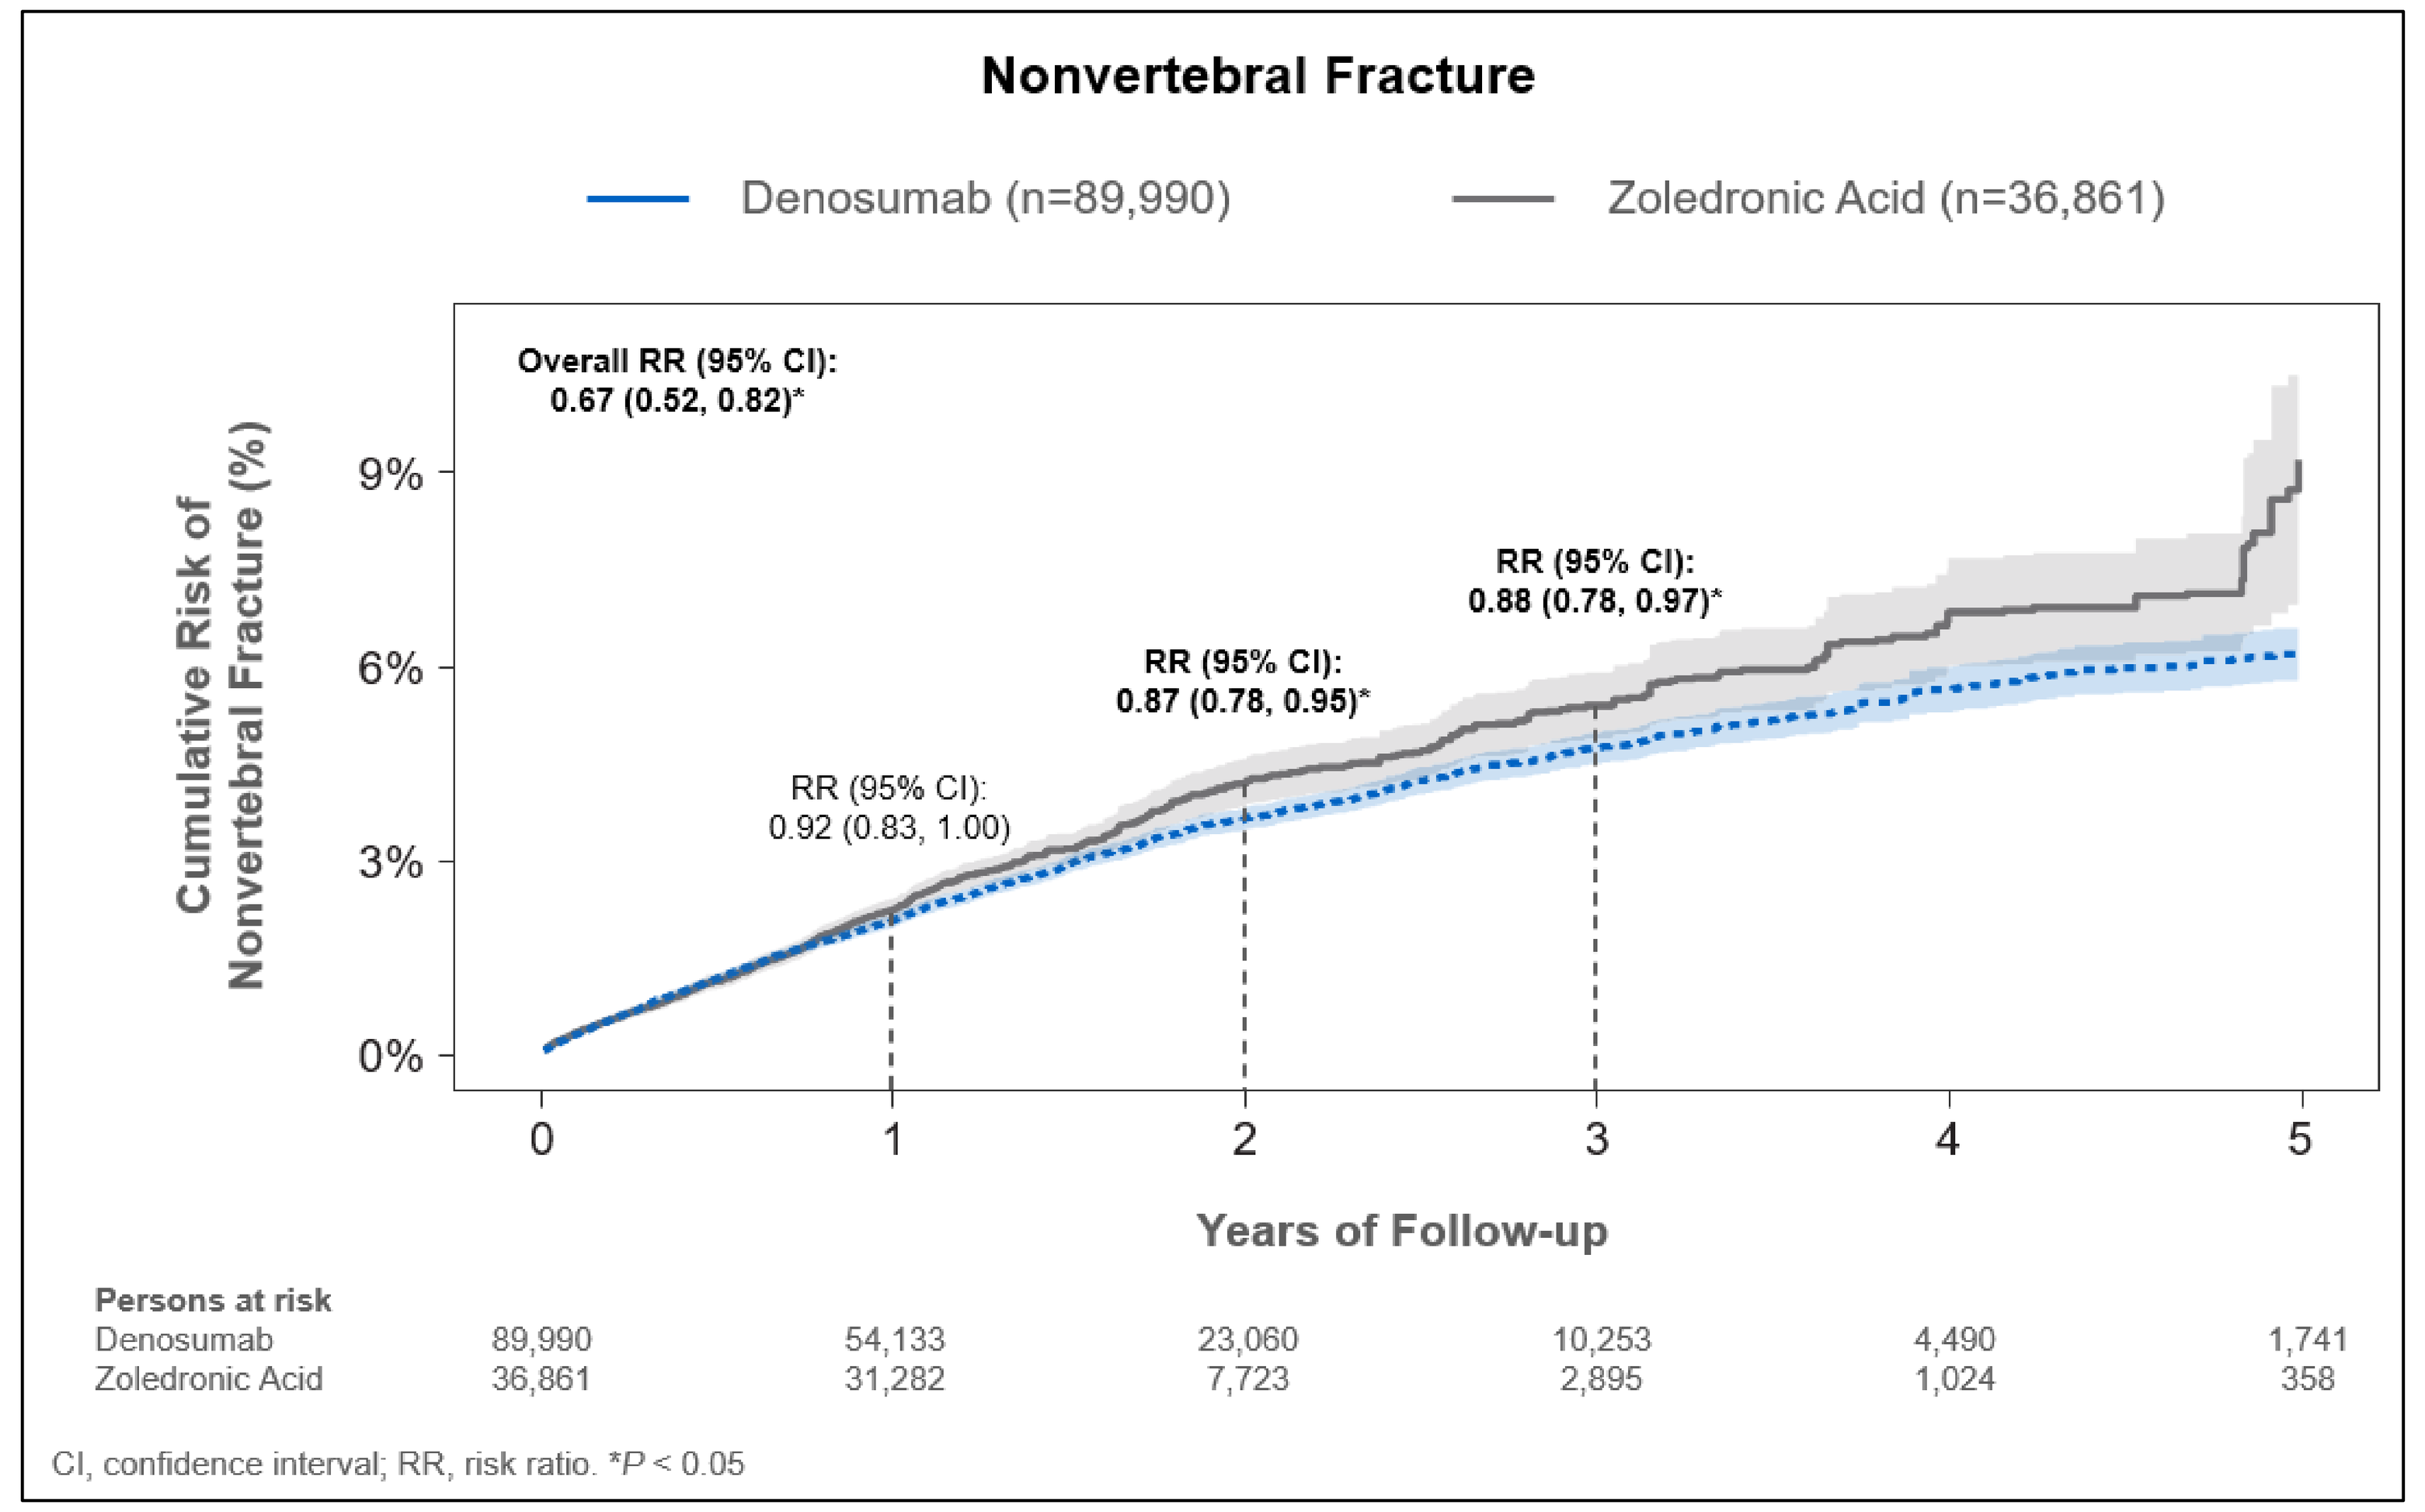
Nonvertebral Fracture
Denosumab (n=89,990)
Zoledronic Acid (n=36,861)
Cumulative Risk of Nonvertebral Fracture (%)
Overall RR (95% CI): 0.67 (0.52, 0.82)*
RR (95% CI): 0.92 (0.83, 1.00)
RR (95% CI): 0.87 (0.78, 0.95)*
RR (95% CI): 0.88 (0.78, 0.97)*
Years of Follow-up
Persons at risk
Denosumab 89,990 54,133 23,060 10,253 4,490 1,741
Zoledronic Acid 36,861 31,282 7,723 2,895 1,024 358
CI, confidence interval; RR, risk ratio. *P < 0.05

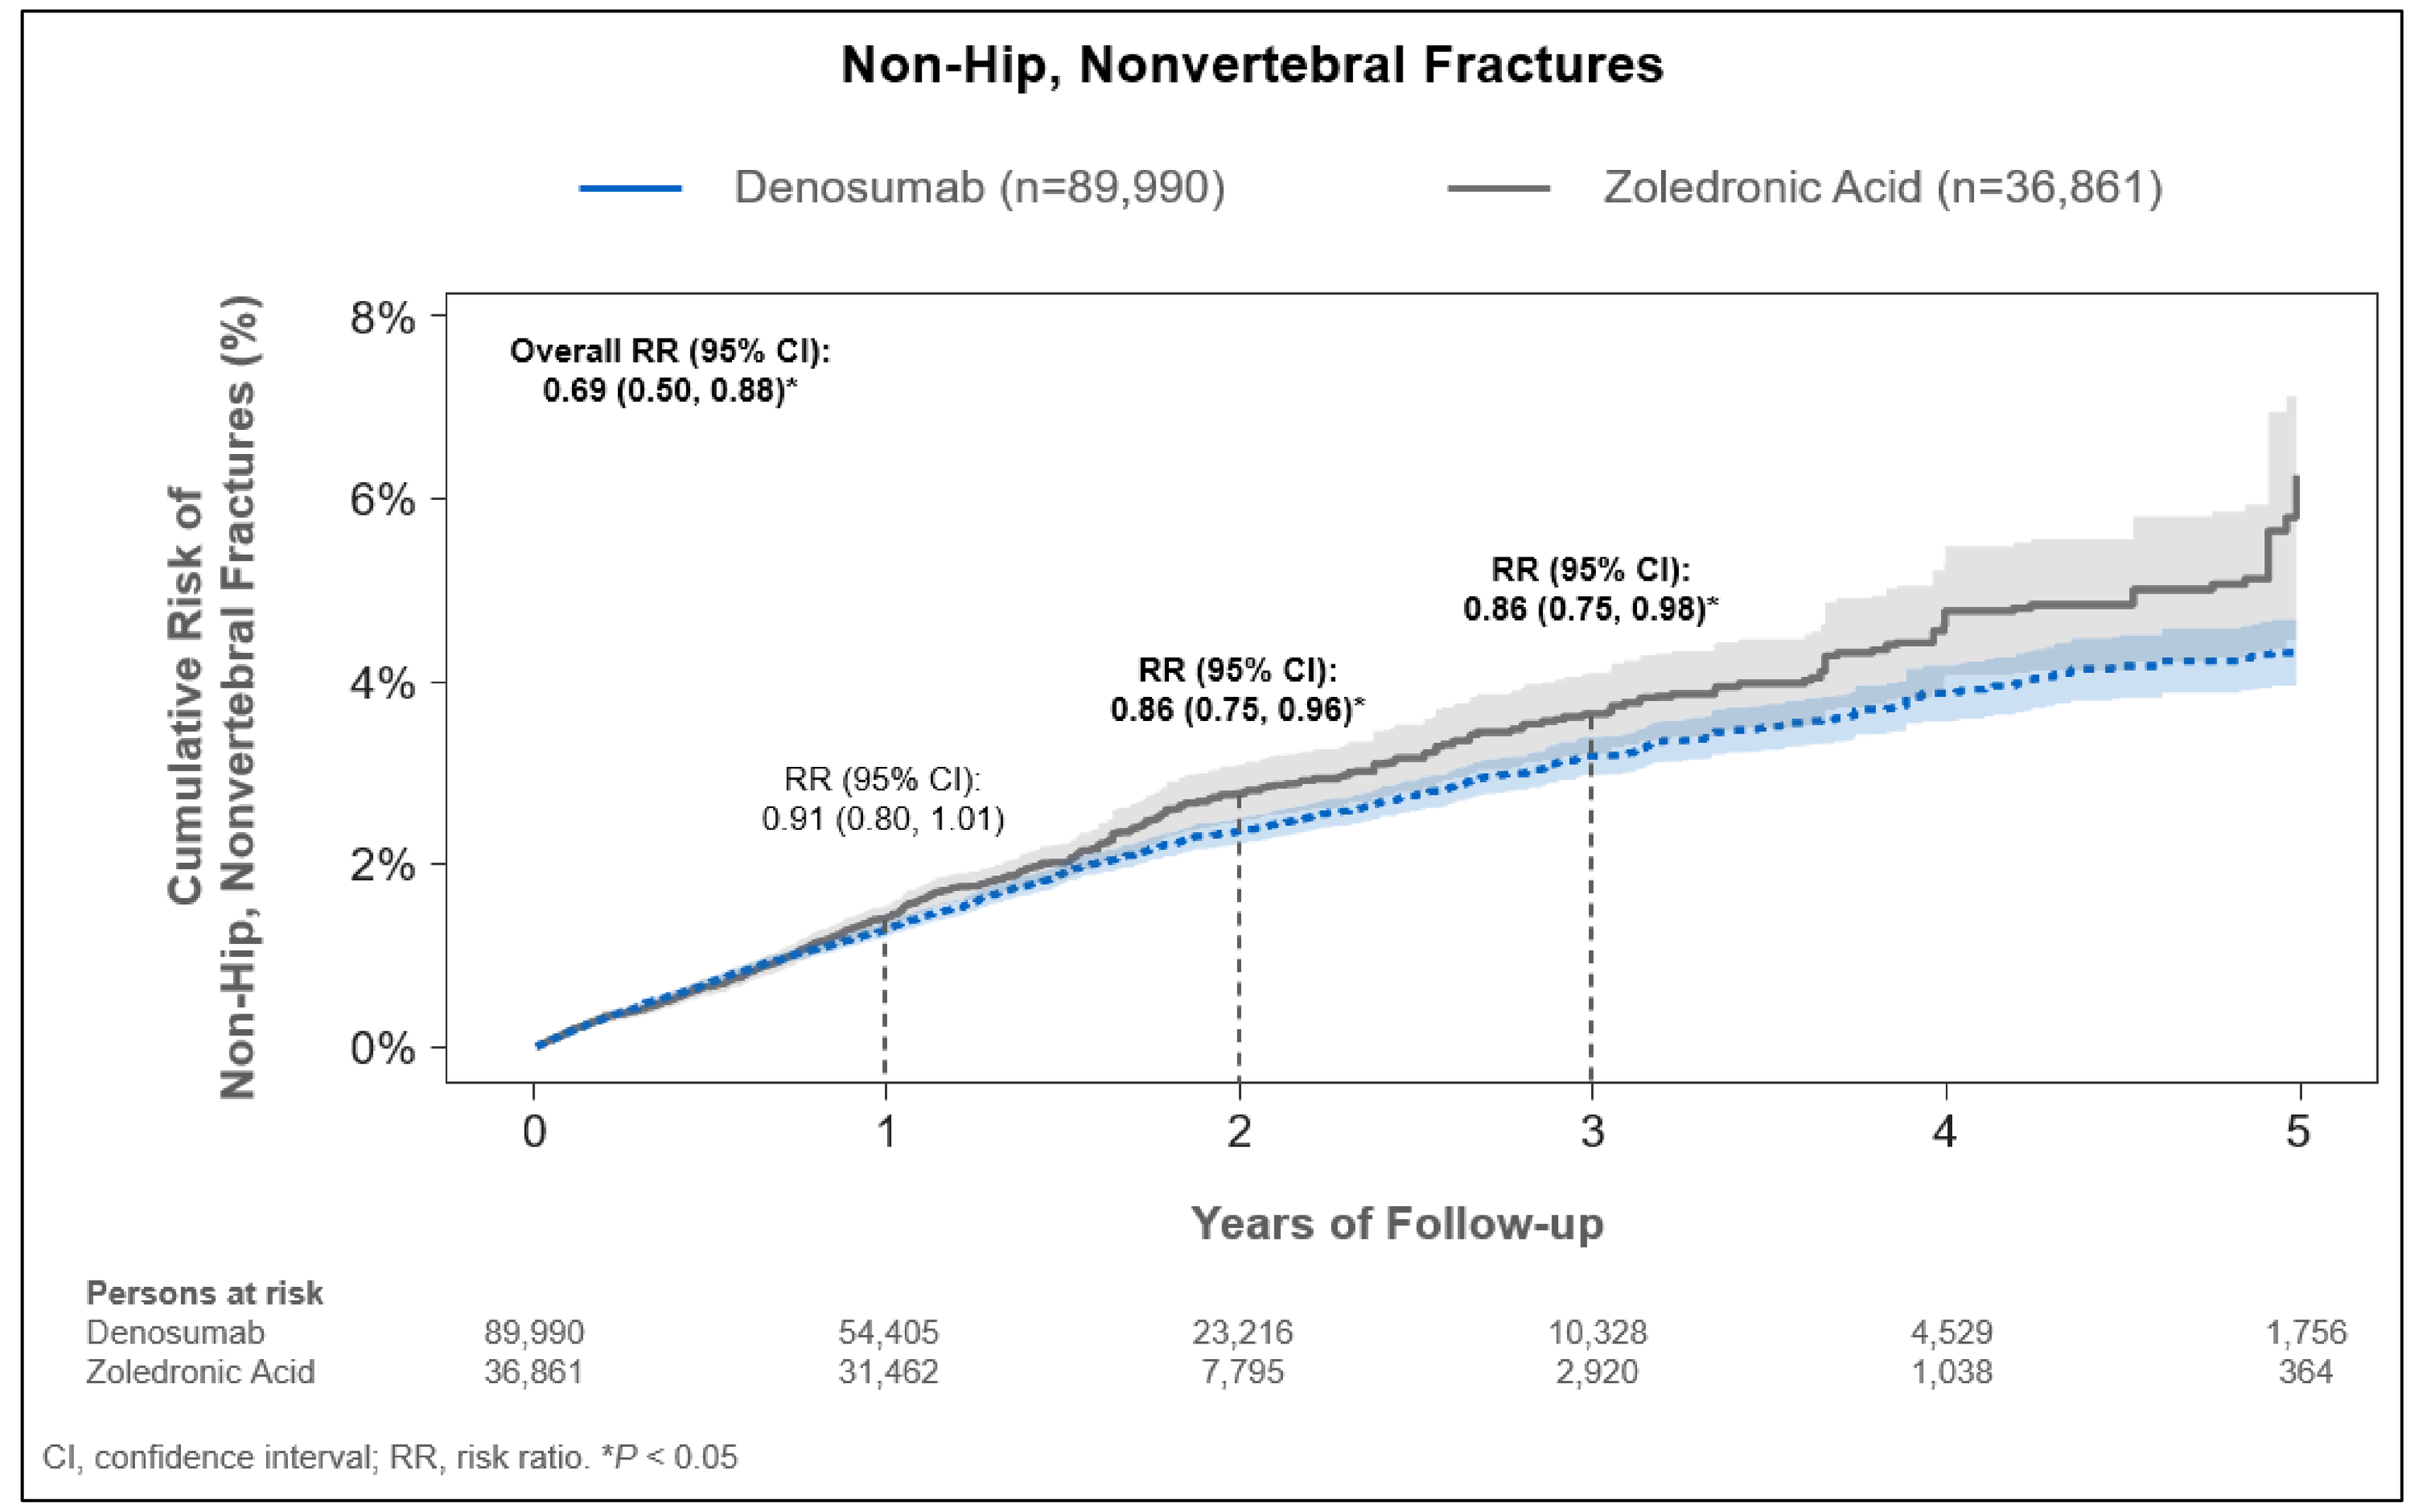
Non-Hip, Nonvertebral Fractures
Denosumab (n=89,990)
Zoledronic Acid (n=36,861)
Cumulative Risk of Non-Hip, Nonvertebral Fractures (%)
Overall RR (95% CI): 0.69 (0.50, 0.88)*
RR (95% CI): 0.91 (0.80, 1.01)
RR (95% CI): 0.86 (0.75, 0.96)*
RR (95% CI): 0.86 (0.75, 0.98)*
Years of Follow-up
Persons at risk
Denosumab 89,990 54,405 23,216 10,328 4,529 1,756
Zoledronic Acid 36,861 31,462 7,795 2,920 1,038 364
CI, confidence interval; RR, risk ratio. *P < 0.05

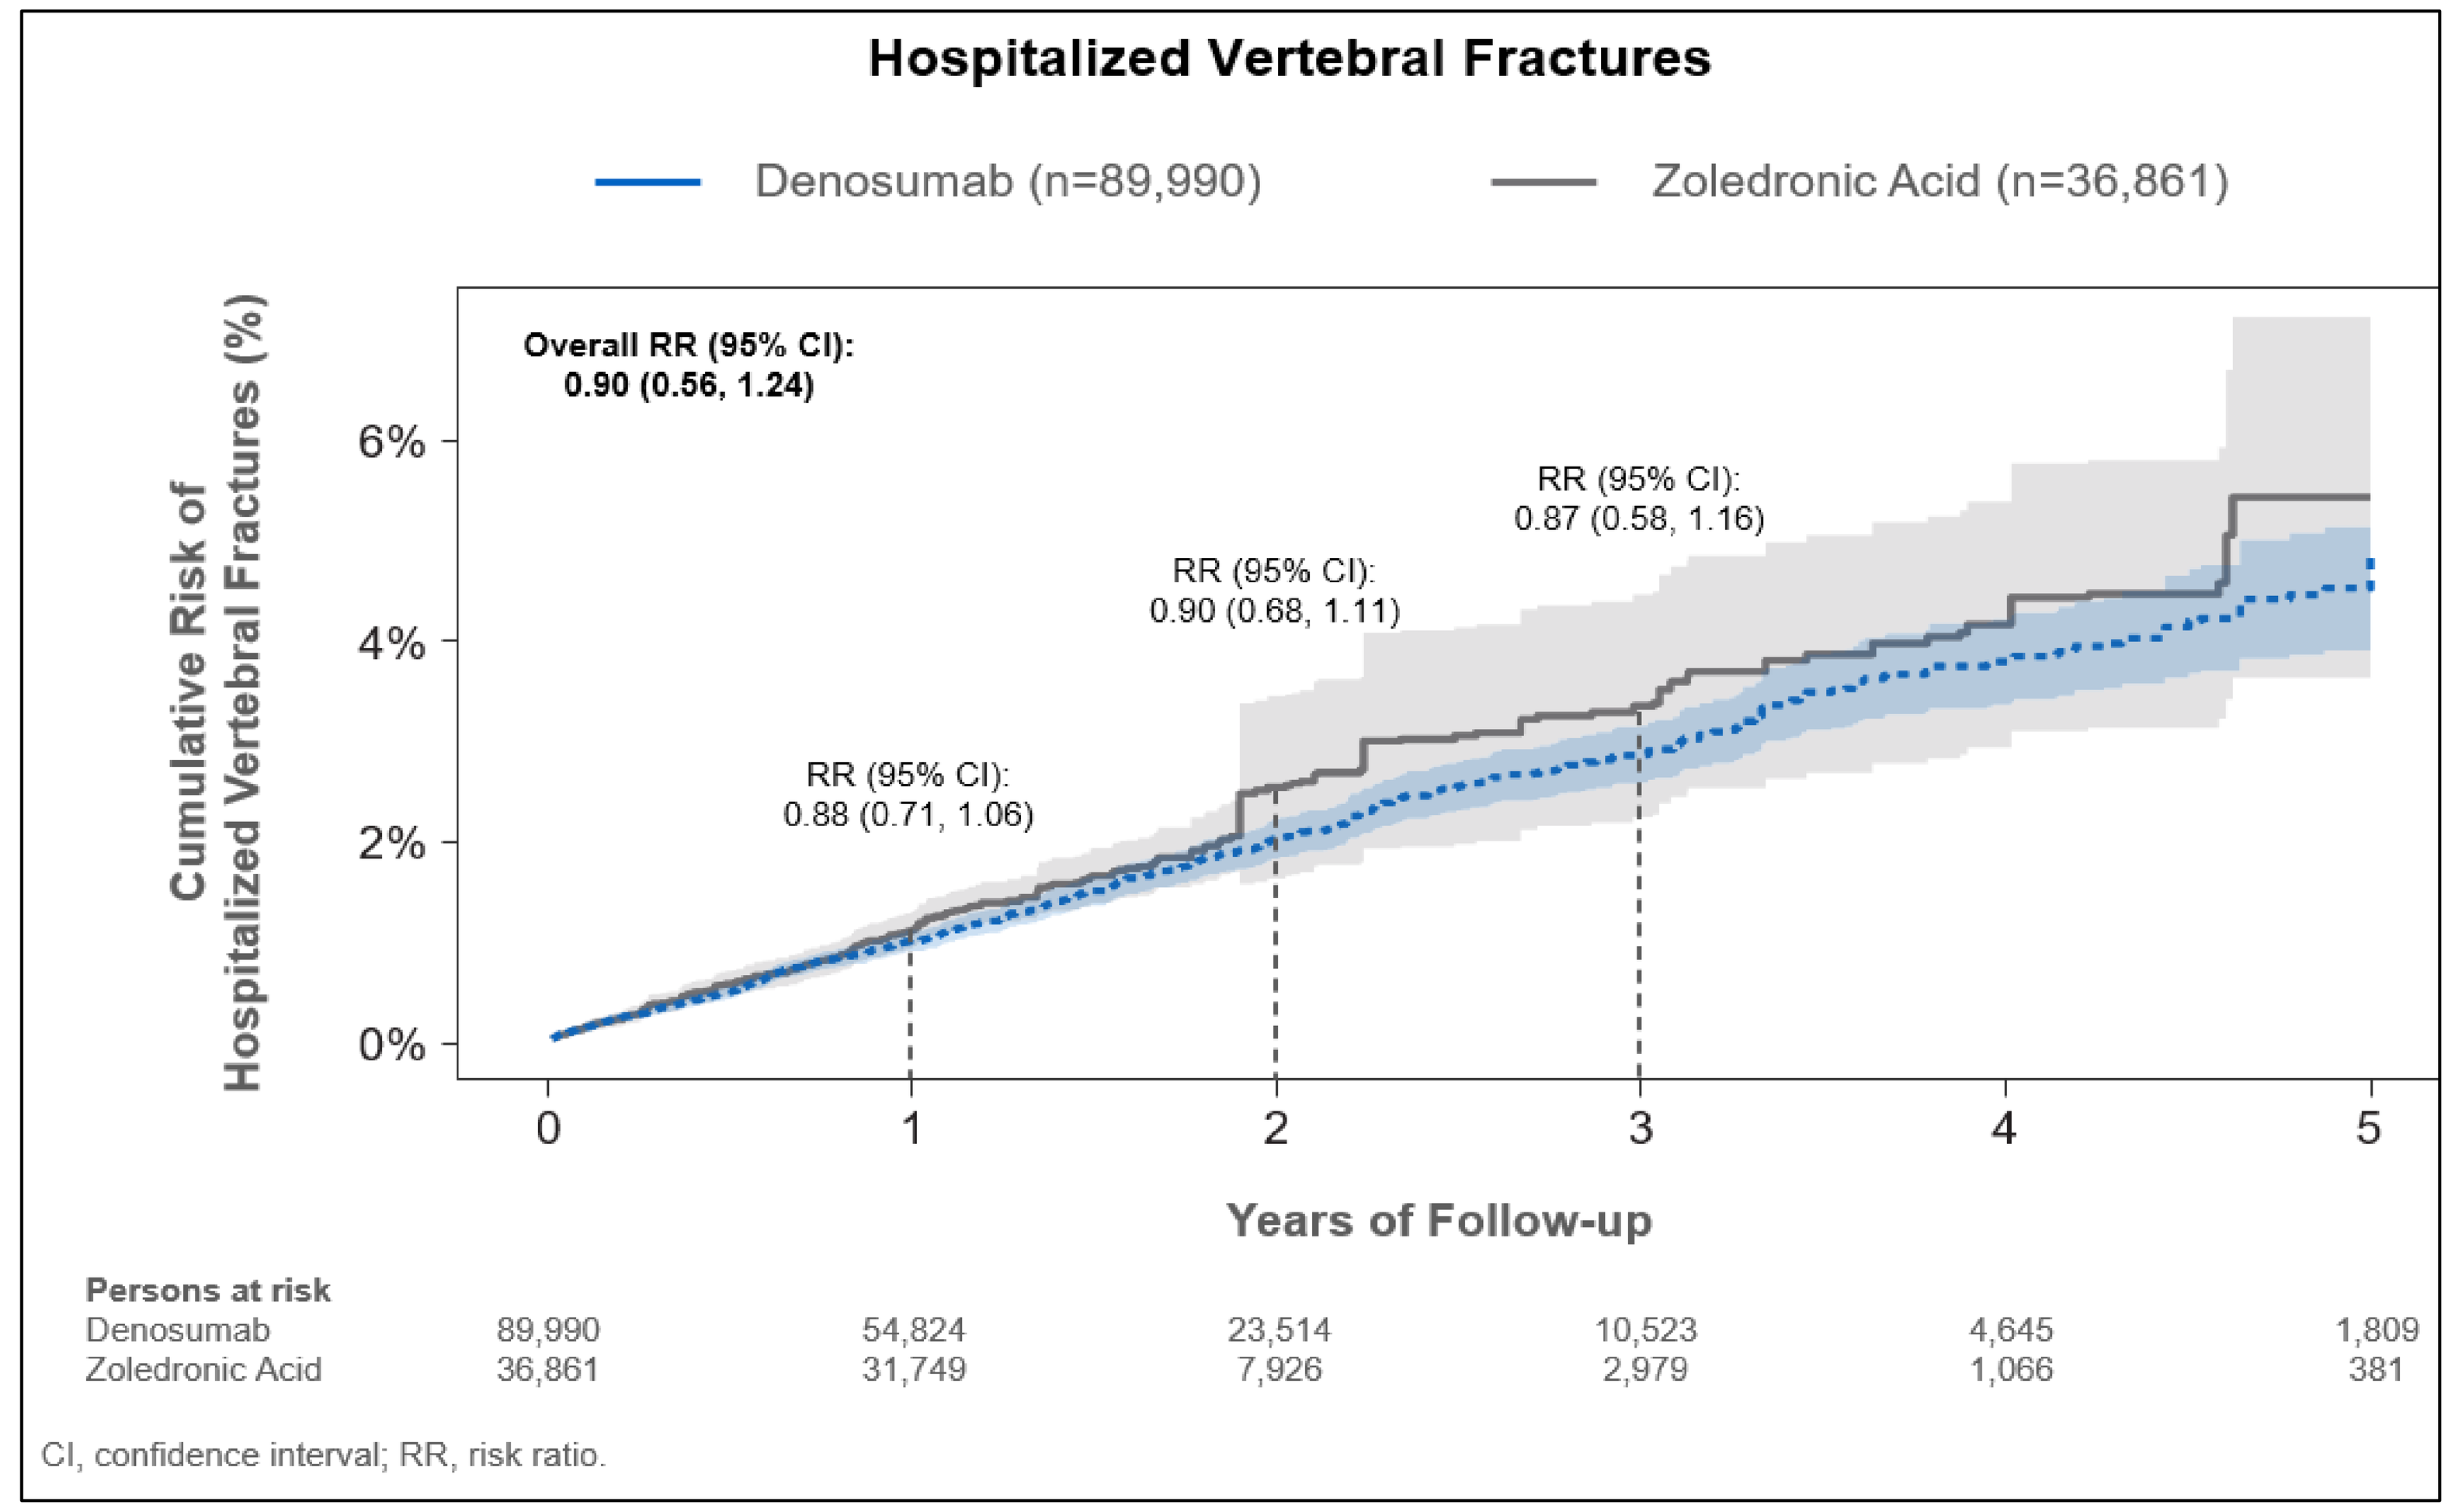
Hospitalized Vertebral Fractures
Denosumab (n=89,990)
Zoledronic Acid (n=36,861)
Cumulative Risk of Hospitalized Vertebral Fractures (%)
Overall RR (95% CI): 0.90 (0.56, 1.24)
RR (95% CI): 0.88 (0.71, 1.06)
RR (95% CI): 0.90 (0.68, 1.11)
RR (95% CI): 0.87 (0.58, 1.16)
Years of Follow-up
Persons at risk
Denosumab 89,990 54,824 23,514 10,523 4,645 1,809
Zoledronic Acid 36,861 31,749 7,926 2,979 1,066 381
CI, confidence interval; RR, risk ratio.

CONCLUSIONS

In a cohort of over 125,000 treatment-naive women with PMO, we observed robust, clinically meaningful reductions in the risk of MOP, hip, NV, and NHNV fractures for patients on Dmab compared to ZA, with greater reductions in fracture risk with longer duration of exposure.