Efficacy and Safety of Lebrikizumab in Adult and Adolescent Patients With Skin of Color and Moderate-to-Severe Atopic Dermatitis: An Interim Analysis of the Open-Label Phase 3b Trial, ADmirable

Andrew Alexis,[1] Ali Moiin,[2] Jill Waibel,[3] Paul Wallace,[4] David Cohen,[5] Pearl Kwong,[6] Amber Reck Atwater,[7] Cynthia Harris,[7] Jennifer Proper,[7] Evangeline Pierce,[7] Angela Moore[8], Sonya Raikar (Non-author presenter)[7]

[1]Weill Cornell Medicine, New York, NY, USA; [2]Comprehensive Dermatology Center, Detroit, MI, USA; [3]Miami Dermatology and Laser Institute, Miami, FL, USA; [4]Wallace Skin Research Center, Los Angeles, CA, USA; [5]Skin Care Physicians of Georgia, Macon, GA, USA; [6]Suncoast Skin Solutions, Inc., Jacksonville, FL, USA; [7]Eli Lilly and Company, Indianapolis, IN, USA; [8]Baylor University Medical Center, Dallas, TX, USA; Arlington Research Center, Arlington, TX, USA; Arlington Center for Dermatology, Arlington, TX, USA

BACKGROUND

- Lebrikizumab is a novel monoclonal antibody that binds with high affinity and slow off-rate to IL-13, thereby blocking the downstream effects of IL-13 with high potency[1]
- The efficacy and safety of lebrikizumab to treat moderate-to-severe AD have been established in Phase 3 studies, including subset analyses by race and ethnicity[2-8]
- There is a paucity of data to guide the treatment of moderate-to-severe AD in populations traditionally under-represented in clinical trials, including patients with skin of color[9]

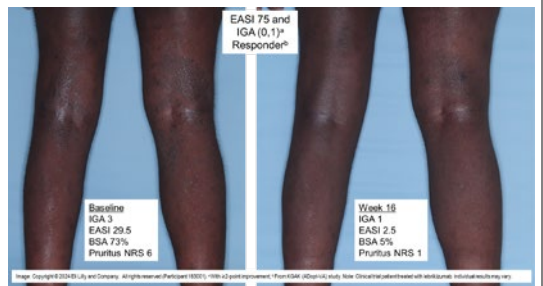
EASI 75 and IGA (0,1)[a] Responder[b]
Baseline
IGA 3
EASI 29.5
BSA 73%
Pruritus NRS 6
Week 16
IGA 1
EASI 2.5
BSA 5%
Pruritus NRS 1

OBJECTIVE

- To present interim 16-week results from Admirable (NCT05372419), the first Phase 3b, open-label, 24-week study to evaluate the safety and efficacy of lebrikizumab in adult and adolescent patients with skin of color and moderate-to-severe AD
- To describe innovative objective measures of pigment, erythema, and post-inflammatory hyper- and hypo-pigmentation

KEY FINDINGS

EASI Responses From Week 0 to Week 16 (N=50)

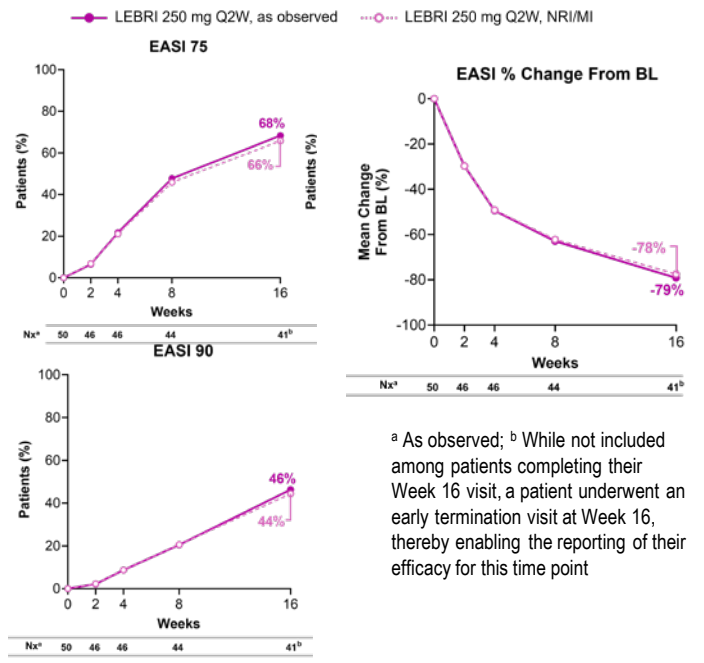
LEBRI 250 mg Q2W, as observed
LEBRI 250 mg Q2W, NRI/MI
EASI 75
68%
66%
EASI 90
46%
44%
EASI % Change From BL
Mean Change From BL (%)
-78%
-79%
Patients (%)
Weeks
Nx[a] 50 46 46 44 41[b]

[a] As observed; [b] While not included among patients completing their Week 16 visit, a patient underwent an early termination visit at Week 16, thereby enabling the reporting of their efficacy for this time point

IGA (0,1) With ≥2-Point Improvement From Week 0 to Week 16 (N=50)

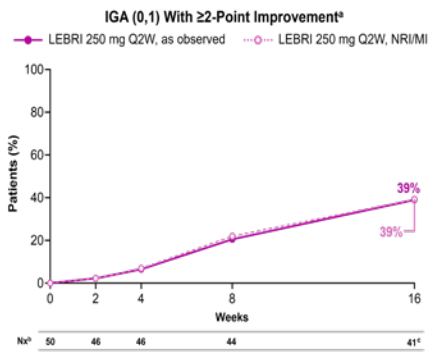
IGA (0,1) With ≥2-Point Improvement[a]
LEBRI 250 mg Q2W, as observed
LEBRI 250 mg Q2W, NRI/MI
39%
39%
Patients (%)
Weeks
Nx[b] 50 46 46 44 41[c]

[a] ITT population with baseline IGA ≥2; [b] As observed; [c] While not included among patients completing their Week 16 visit, a patient underwent an early termination visit at Week 16, thereby enabling the reporting of their efficacy for this time point

Pruritus NRS With ≥4- and ≥3-Point Improvement Response From Week 0 to Week 16

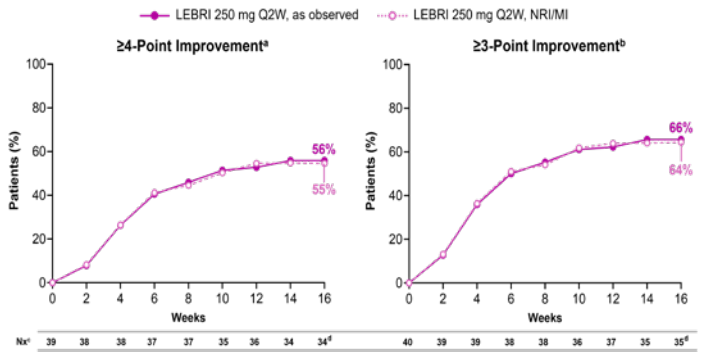
LEBRI 250 mg Q2W, as observed
LEBRI 250 mg Q2W, NRI/MI
≥4-Point Improvement[a]
56%
55%
≥3-Point Improvement[b]
66%
64%
Patients (%)
Weeks
Nx[c] 39 38 38 37 37 35 36 34 34[d]
40 39 39 38 38 36 37 35 35[d]

[a] ITT population with baseline Pruritus NRS ≥4 (N=39); [b] ITT population with baseline Pruritus NRS ≥3 (N=40); [c] As observed; [d] While not included among patients completing their Week 16 visit, a patient underwent an early termination visit at Week 16, thereby enabling the reporting of their efficacy for this time point

CONCLUSIONS

- This is the first Phase 3b clinical trial of lebrikizumab for patients with moderate-to-severe AD
 - With skin of color (80% Black or African American) and Fitzpatrick Phototype IV-VI
 - Using objective and subjective tools and scales to evaluate signs and symptoms that matter to patients
- Lebrikizumab improved AD signs and symptoms in patients with skin of color and moderate-to-severe AD after 16 weeks of treatment, with
 - EASI 75 achieved in 68% of patients
 - IGA (0,1) with ≥2-point improvement achieved in 39% of patients
 - Pruritus NRS ≥4-point improvement achieved in 56% of patients
 - No serious adverse events[a]
- The innovative objective measure PDCA-Derm™[,b] identified improvement in post-inflammatory hyperpigmented lesions in 12/21 patients and improvement to normal skin tone in 6/21 patients at Week 16

[a] Results from interim analysis; full data will be reported when study is complete; [b] A scale used to compare post-inflammatory lesions to unaffected adjacent normal skin

METHODS

Study Design – Interim Analysis

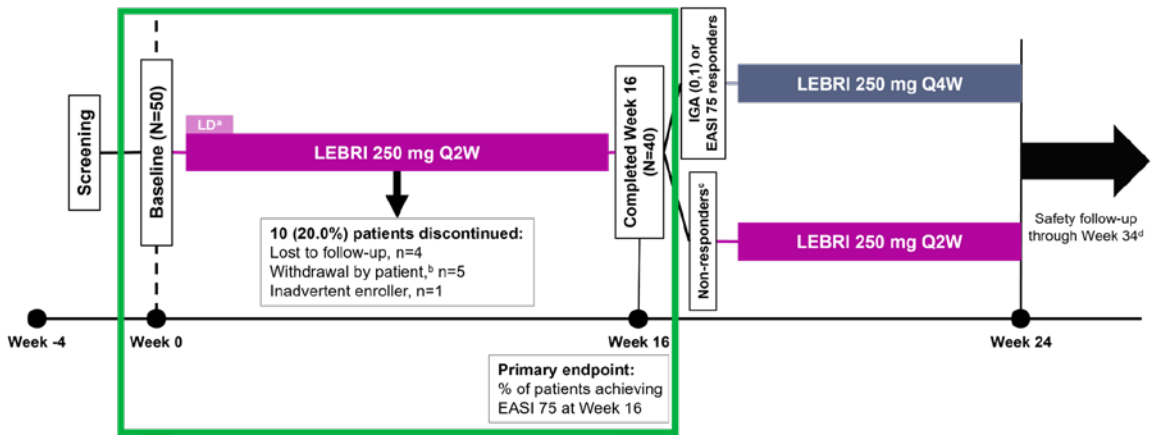
Screening
Baseline (N=50)
LD[a]
LEBRI 250 mg Q2W
10 (20.0%) patients discontinued:
Lost to follow-up, n=4
Withdrawal by patient,[b] n=5
Inadvertent enroller, n=1
Completed Week 16 (N=40)
IGA (0,1) or EASI 75 responders
Non-responders[c]
LEBRI 250 mg Q4W
LEBRI 250 mg Q2W
Safety follow-up through Week 34[d]
Week -4
Week 0
Week 16
Week 24
Primary endpoint:
% of patients achieving EASI 75 at Week 16

[a] 500-mg LD at Weeks 0 and 2; [b] Reasons included personal issue unrelated to trial; expressed disinterest when asked to get additional hepatitis testing; no longer wants to take IP injections; withdrew consent due to low calcium levels; eczema resolved; [c] Did not achieve IGA (0,1) or EASI 75; [d] Safety follow-up approximately 12 weeks after last study injection
Note: The use of low- and/or mid-potency TCS, TCIs, topical PDE-4 inhibitors, or high-potency TCS up to 10 days was permitted. Patients requiring rescue therapy (high-potency TCS >10 days, topical JAK inhibitors, phototherapy, systemic medication) were discontinued from the study

Outcomes and Statistical Analysis

Week 16 Outcomes

- EASI,[a,b] IGA (0,1),[b,c] and Pruritus NRS[d]
- PDCA-Derm™[,e]
- PO-SCORAD, Mexameter (melanin), Scarletred (erythema), biomarkers, and microbiome[f]

Statistical Analyses

- Interim analysis includes patients who enrolled by June 29, 2023, and completed 16 weeks of lebrikizumab therapy or discontinued treatment by Week 16
- *Primary analysis:* All data were summarized as observed
- *Supplemental analysis:* Missing data due to lack of efficacy were imputed with NRI; other missing data were imputed with MI

[a] A composite index with scores ranging from 0 to 72, with higher values indicating more severe and/or extensive disease; [b] Investigators received training on accurate assessment of EASI and IGA for patients with skin of color; [c] A 5-point scale that provides a global clinical assessment of AD severity ranging from 0 to 4, where 0 indicates clear, 2 is mild, 3 is moderate, and 4 indicates severe AD; [d] A patient-reported, single-item, 11-point scale that is used daily to rate worst itch severity over the past 24 hours (0 indicates "no itch"; 10 indicates "worst itch imaginable"); [e] A scale used to compare post-inflammatory lesions to unaffected, adjacent normal skin; [f] These outcomes are not included in this analysis

Key Eligibility Criteria

- Adults (≥18 years) and adolescents (≥12 to <18 years; weight ≥40 kg)
- Self-reported race, including Black or African American, American Indian or Alaska Native, Asian, and Native Hawaiian or Other Pacific Islander
- Fitzpatrick Phototype IV, V, or VI
- Chronic AD (according to AAD Consensus Criteria[10]) for ≥1 year
- Moderate-to-severe AD, including baseline:
 - EASI ≥16
 - IGA ≥3
 - BSA involvement ≥10%
- History of inadequate response to topical medications
- Naive to biologics indicated for the treatment of AD

RESULTS

Baseline Demographics

	LEBRI 250 mg Q2W (N=50)
Age, years	42.2 (19.7)
Adult (≥18 years), n (%)	42 (84.0)
Adolescent (≥12 to <18 years), n (%)	8 (16.0)
Female, n (%)	23 (46.0)
BMI, kg/m^2	30.2 (7.7)
BMI category, n (%)	
Underweight (<18.5 kg/m^2)	3 (6.0)
Normal (≥18.5 and <25 kg/m^2)	8 (16.0)
Overweight (≥25 and <30 kg/m^2)	15 (30.0)
Obese (≥30 and <40 kg/m^2)	17 (34.0)
Extreme obese (≥40 kg/m^2)	7 (14.0)
Age at AD onset, years	22.8 (22.8)

Note: Data are mean (SD) unless stated otherwise

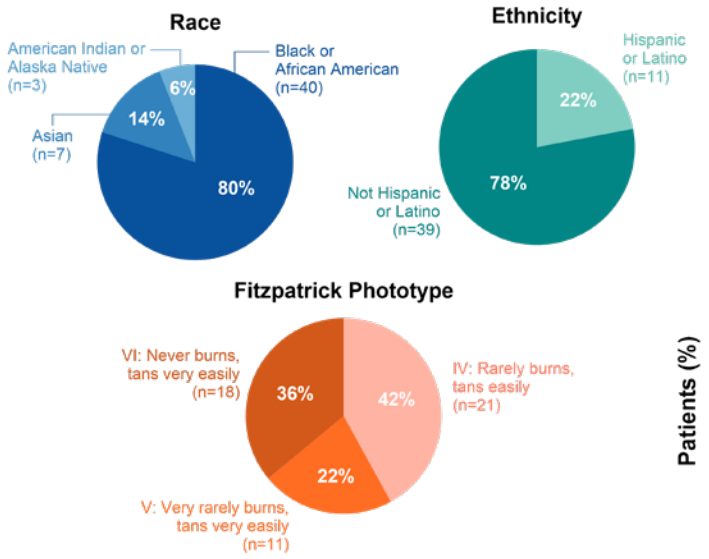
Race
American Indian or Alaska Native (n=3)
Asian (n=7)
Black or African American (n=40)
6%
14%
80%
Ethnicity
Hispanic or Latino (n=11)
Not Hispanic or Latino (n=39)
22%
78%
Fitzpatrick Phototype
VI: Never burns, tans very easily (n=18)
IV: Rarely burns, tans easily (n=21)
V: Very rarely burns, tans very easily (n=11)
36%
42%
22%

Baseline Disease Characteristics

	LEBRI 250 mg Q2W (N=50)
IGA[a]	
3 (Moderate), n (%)	32 (64.0)
4 (Severe), n (%)	17 (34.0)
EASI	28.1 (12.4)
BSA % affected	41.7 (20.8)
Pruritus NRS[b]	7.2 (2.2)
≥3, n (%)	40 (93.0)
≥4, n (%)	39 (90.7)
Sleep-Loss scale[b]	2.1 (1.1)
DLQI[c]	13.9 (8.3)
CDLQI[c]	13.0 (9.6)
PO-SCORAD[d]	57.5 (18.7)
PDCA-Derm™	
Hypopigmented lesion(s), n (%)	9 (18.0)
Hyperpigmented lesion(s), n (%)	27 (54.0)

[a] 1 patient inadvertently enrolled with IGA=2 and discontinued when discovered they did not meet enrollment criteria; [b] Nx=43; [c] Patients <16 years of age at baseline completed CDLQI [Nx=6]; others completed DLQI [Nx=42]; [d] Nx=48
Note: Data are mean (SD) unless stated otherwise. At baseline, patients could have hyperpigmented lesions, hypopigmented lesions, both, or neither.

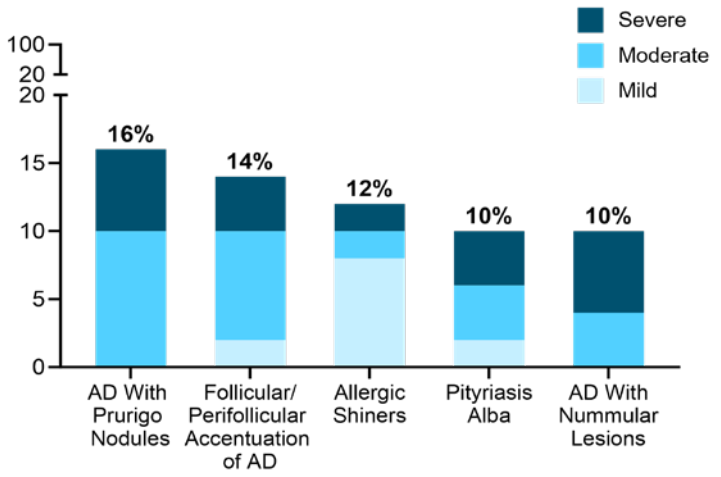
Severe
Moderate
Mild
Patients (%)
16%
14%
12%
10%
10%
AD With Prurigo Nodules
Follicular/ Perifollicular Accentuation of AD
Allergic Shiners
Pityriasis Alba
AD With Nummular Lesions

PDCA-Derm™: A Scale Used to Compare Post-inflammatory Lesions To Unaffected Adjacent Normal Skin[1]

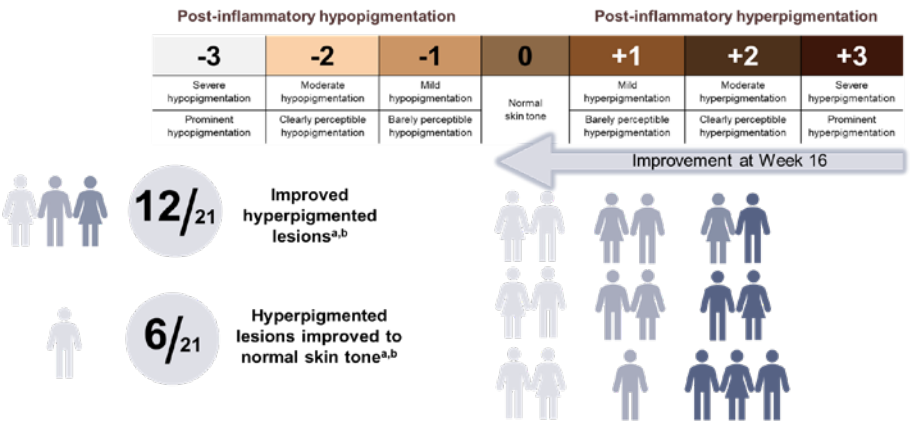
Post-inflammatory hypopigmentation
Post-inflammatory hyperpigmentation
-3 Severe hypopigmentation Prominent hypopigmentation
-2 Moderate hypopigmentation Clearly perceptible hypopigmentation
-1 Mild hypopigmentation Barely perceptible hypopigmentation
0 Normal skin tone
+1 Mild hyperpigmentation Barely perceptible hyperpigmentation
+2 Moderate hyperpigmentation Clearly perceptible hyperpigmentation
+3 Severe hyperpigmentation Prominent hyperpigmentation
Improvement at Week 16
12/21 Improved hyperpigmented lesions[a,b]
6/21 Hyperpigmented lesions improved to normal skin tone[a,b]

[a] Data as observed; [b] Three patients whose PDCA-Derm™ went from +2 to -2 and +2 to -1 not shown in figure
Notes: 1) The colors in the PDCA-Derm™ table serve as visual aids for comprehension and are not indicative of actual skin tones; 2) For patients with multiple hypopigmented or hyperpigmented lesions at baseline, only the lesion with the most severe score was included in the analysis for each lesion type. In the event of a tie, the lesion reflecting a smaller improvement or worsening in condition from baseline to Week 16 was included. 3) Images do not represent actual gender breakdown in study.

Use of Concomitant Topical and Systemic Therapy

	LEBRI 250 mg Q2W (N=50)
Topical therapy	
Low-potency TCS	14 (28.0)
Mid-potency TCS	13 (26.0)
High-potency TCS	0
High-potency TCS >10 days	0
TCIs	7 (14)
Topical PDE-4 inhibitor	7 (14)
Topical JAK inhibitor	0
Systemic therapy	
Prednisone	1 (2)
Immunosuppressant	0
Biologic	0
Phototherapy or photochemotherapy	0

Notes: Data are n (%). Rescue therapy is defined as high-potency TCS >10 days, topical JAK inhibitors, phototherapy, or systemic medication

REFERENCES

1. Okragly AJ, et al. *Dermatol Ther (Heidelb)*. 2023;13:1535-1547.
2. Silverberg JI, et al. *N Engl J Med*. 2023;388:1080-1091.
3. Blauvelt A, et al. *Br J Dermatol*. 2023;188:740-748.
4. Simpson EL, et al. *JAMA Dermatol*. 2023;159:182-191.
5. Paller AS, et al. *Dermatol Ther (Heidelb)*. 2023;13:1517-1534.
6. Stein Gold L, et al. *Am J Clin Dermatol*. 2023;24:595-607.
7. Alexis A, et al. Poster presented at: *RAD Virtual 2023. Poster 115*
8. Soung J, et al. Poster presented at: *ISID 2023* (ADopt-VA). Poster LB1702.
9. Chen V, et al. *JAMA Dermatol*. 2022;158:288-292.
10. Eichenfield LF, et al. *J Am Acad Dermatol*. 2014;70:338-351.

ABBREVIATIONS

AAD=American Academy of Dermatology; AD=Atopic Dermatitis; BSA=Body Surface Area; CDLQI=Children's Dermatology Life Quality Index; DLQI = Dermatology Life Quality Index; EASI 75=75% improvement from baseline in eczema area and severity index; EASI=Eczema Area and Severity Index; IGA (0,1)=investigator's global assessment response of clear or almost clear; IP=investigational product; IGA (0,1)=investigator's Global Assessment Response of Clear or Almost Clear; ITT= Intent to Treat; JAK=Janus Kinase; Lebri=Lebrikizumab; LD=Loading dose; Mi=multiple Imputation; NRI=Non-Responder Imputation; NRS=numeric Rating Scale; Nx = number of patients with non-missing values; PDE-4=Phosphodiesterase 4; PO-SCORAD=Patient-Oriented Scoring of Atopic Dermatitis Q2W=every 2 weeks; Q4W=every 4 weeks; SD= Standard Deviation; TCI=Topical Calcineurin Inhibitor; TCS=Topical Corticosteroids

DISCLOSURES

A. Alexis has received grant support from: AbbVie, Amgen, Arcutis, Castle Biosciences, Dermavant, Galderma, and LEO Pharma; has served as a consultant and/or advisor for: AbbVie, Allergan, Almirall, Alphyn Biologics, Amgen, Apogee Therapeutics, Arcutis, Bausch Health, Beiersdorf, Bristol Myers Squibb, Canfield Scientific, Cara Therapeutics, Castle Biosciences, Cutera, Dermavant, Eli Lilly and Company, EPI Health, Incyte Corporation, Galderma, Janssen, LEO Pharma, L'Oréal, Ortho Dermatologics, Pfizer, Sanofi Regeneron, Swiss American CDMO, UCB Pharma, VisualDx, and VYNE Therapeutics; has served as a speaker for: Bristol Myers Squibb, Janssen, Johnson & Johnson, L'Oréal, Regeneron, and Sanofi Genzyme; has received royalties from: Springer Science+Business Media, Wiley-Blackwell, and Wolters Kluwer Health; **A. Moiin** and **D. Cohen** have no conflicts of interest; **J. Waibel** has been a consultant and/or investigator for and/or has received personal fees from: Allergan, Aquavit Pharmaceuticals, Argenx, BellaMia Technologies, Bristol Myers Squibb, Candela Healthcare, Cytrellis Biosystems, Eli Lilly and Company, Galderma, Horizon Therapeutics, Incyte Corporation, Lutris Pharma, OliX Pharmaceuticals, P&G Pharmaceuticals, Pfizer, RegenX Science, Sanofi, Shanghai Pharma Biotherapeutics, SkinCeuticals, and Turn Biotechnologies; **P. Wallace** has served as a principal investigator, advisor, and/or speaker for: AbbVie, Amgen, Arcutis, Biogen, Bristol Myers Squibb, Celgene, Centocor, Cynosure, Eli Lilly and Company, Genentech, GlaxoSmithKline, Merck, Novartis, Pfizer, Sanofi, Sanofi-Aventis, and UCB Pharma; **P. Kwong** is a principal investigator for: AbbVie, Amgen, Arcutis, Castle Creek Biosciences, Dermavant, Eli Lilly and Company, Galderma, Incyte Corporation, Novartis, Pfizer, and UCB Pharma; a consultant/advisor for: Bristol Myers Squibb, Eli Lilly and Company, EPI Health, Galderma, Incyte Corporation, LEO Pharma, Novan, Ortho Dermatologics, Pfizer, Sanofi Regeneron, UCB Pharma, and Verrica Pharmaceuticals; and a speaker for: AbbVie, Arcutis, EPI Health, Incyte Corporation, L'Oréal, Ortho Dermatologics, Sanofi Regeneron, Sun Pharma, and Verrica Pharmaceuticals; **A. Reck Atwater**, **C. Harris**, **J. Proper, E. Pierce and**, **G. Gaia** are employees and shareholders of: Eli Lilly and Company; **A. Moore** has served as a consultant, advisory board member, speaker, and/or investigator for: AbbVie, Almirall, Arcutis, Biofrontera, Boehringer Ingelheim, Bristol Myers Squibb, Dermavant, DS Biopharma, Eli Lilly and Company, EPI Health, Evolus, Galderma, Incyte Corporation, Janssen, LEO Pharma, Mayne Pharma, Nimbus Therapeutics, Novartis, PAREXEL, Pfizer, UCB Pharma, Verrica Pharmaceuticals, and VYNE Therapeutics.
Medical writing assistance was provided by Loredana Spoerri, PhD, of ProScribe – Envision Pharma Group, and was funded by Eli Lilly and Company
This study was previously presented at American Academy of Dermatology; San Diego, USA; 8-12 March 2024

These studies, with the exception of ADhere-J, were funded by Dermira, a wholly owned subsidiary of Eli Lilly and Company. ADhere-J was funded by Eli Lilly and Company. Almirall, S.A. has licensed the rights to develop and commercialize lebrikizumab for the treatment of dermatology indications, including atopic dermatitis, in Europe.
Lilly has exclusive rights for development and commercialization of lebrikizumab in the United States and the rest of the world outside of Europe.

Scan the QR code for the audio recording.

Scan the QR code for a list of all Lilly content presented at the congress.

Other company and product names are trademarks of their respective owners.

Copyright © 2024 Eli Lilly and Company and Almirall, S. A. All rights reserved.

National Association of Specialty Pharmacy (NASP); Nashville, TN, USA; October 6-9, 2024